Digestive Disease Week 2021 — Poster #Fr488

Targeted delivery of soluble tofacitinib citrate to the site of inflammation to improve efficacy and safety

Shaoying Nikki Lee, PhD[1]; Cheryl Stork, PhD[1]; Chris Wahl, MD[1]; Sharat Singh, PhD[1]; Emil Chuang, MD[1]
[1]Progenity, Inc., San Diego, CA

progenity®

Introduction

There is an urgent need to achieve higher rates of clinical response, remission, and mucosal healing in inflammatory bowel disease (IBD). The Janus kinase/signal transducers and activators of transcription (JAK/STAT) signaling pathways are important in controlling the aberrant immune response of IBD. Tofacitinib is a potent Janus kinase (JAK) inhibitor with selectivity toward homo- and heterodimers of JAK1 and JAK3, and less potent against JAK2 and JAK2 homodimers *in vitro*.[1] Inhibition of the JAK2 homodimer has been associated with the risk of anemia, neutropenia, thrombocytopenia, infections, lymphoma, and the inhibition of anti-inflammatory responses in the intestine.[2, 3] Tofacitinib was the first oral JAK inhibitor approved to treat moderate to severe ulcerative colitis (UC). During development, a clear dose-response relationship was observed in terms of efficacy, up to 15 mg twice daily (BID), but only the 5 mg and 10 mg BID doses were approved for clinical use in UC due to observed systemic toxicity. Targeted local delivery of drugs to the colon may increase local tissue concentration to improve efficacy and lower systemic absorption. Pharmacokinetics (PK), pharmacodynamics (PD), and biodistribution of tofacitinib liquid formulation through local administration to the cecum were assessed in animal models with surgical implantation of a cecal cannula.

Methods

All animals underwent surgical implantation of a cecal cannula 2 weeks prior dosing. Colitis was induced by exposure to 3% DSS-treated drinking water from Day 0 to Day 5 in male C57BL/6 mice. Dosing was performed at the peak of disease status (Day 12) via intra-cecal catheter (IC) or oral gavage (PO). Animals received a single dose of control vehicle or tofacitinib citrate (Tofa) suspension in carboxyl methyl cellulose (CMC) via PO (15 and 45 mg/kg) or IC delivery (1, 3, & 10 mg/kg). Direct measurement of total and phosphorylated STAT1, 3, & 5 were quantified through immunohistochemistry in colon tissue to compare the PD effects via IC vs. PO.

The biodistribution and coverage of soluble Tofa formulation was evaluated in Yorkshire-Cross Swine, because their gastrointestinal physiology is more similar to humans'. Animals received 4 IC QD doses of Tofa oral suspension (TOF) or Tofa solubilized formulation (TSF) via an implanted intra-cecal catheter at 0.28 mg/kg/dose or 0.46 mg/kg/dose.

Results

Approximately 10- to 15-fold lower doses of Tofa could be used via intra-cecal delivery to achieve equivalent drug concentrations with minimal systemic drug exposure compared to PO.

- Mean plasma concentrations of Tofa in all IC treated groups were ~ 10- to 100-fold lower than the *in vitro* JAK2 IC_{50} (1377 nM) (**Figure 1**).
- At a similar dose level, IC Tofa 10 mg/kg resulted in ~ 20-fold higher tissue/plasma exposure ratio than PO 15 mg/kg.

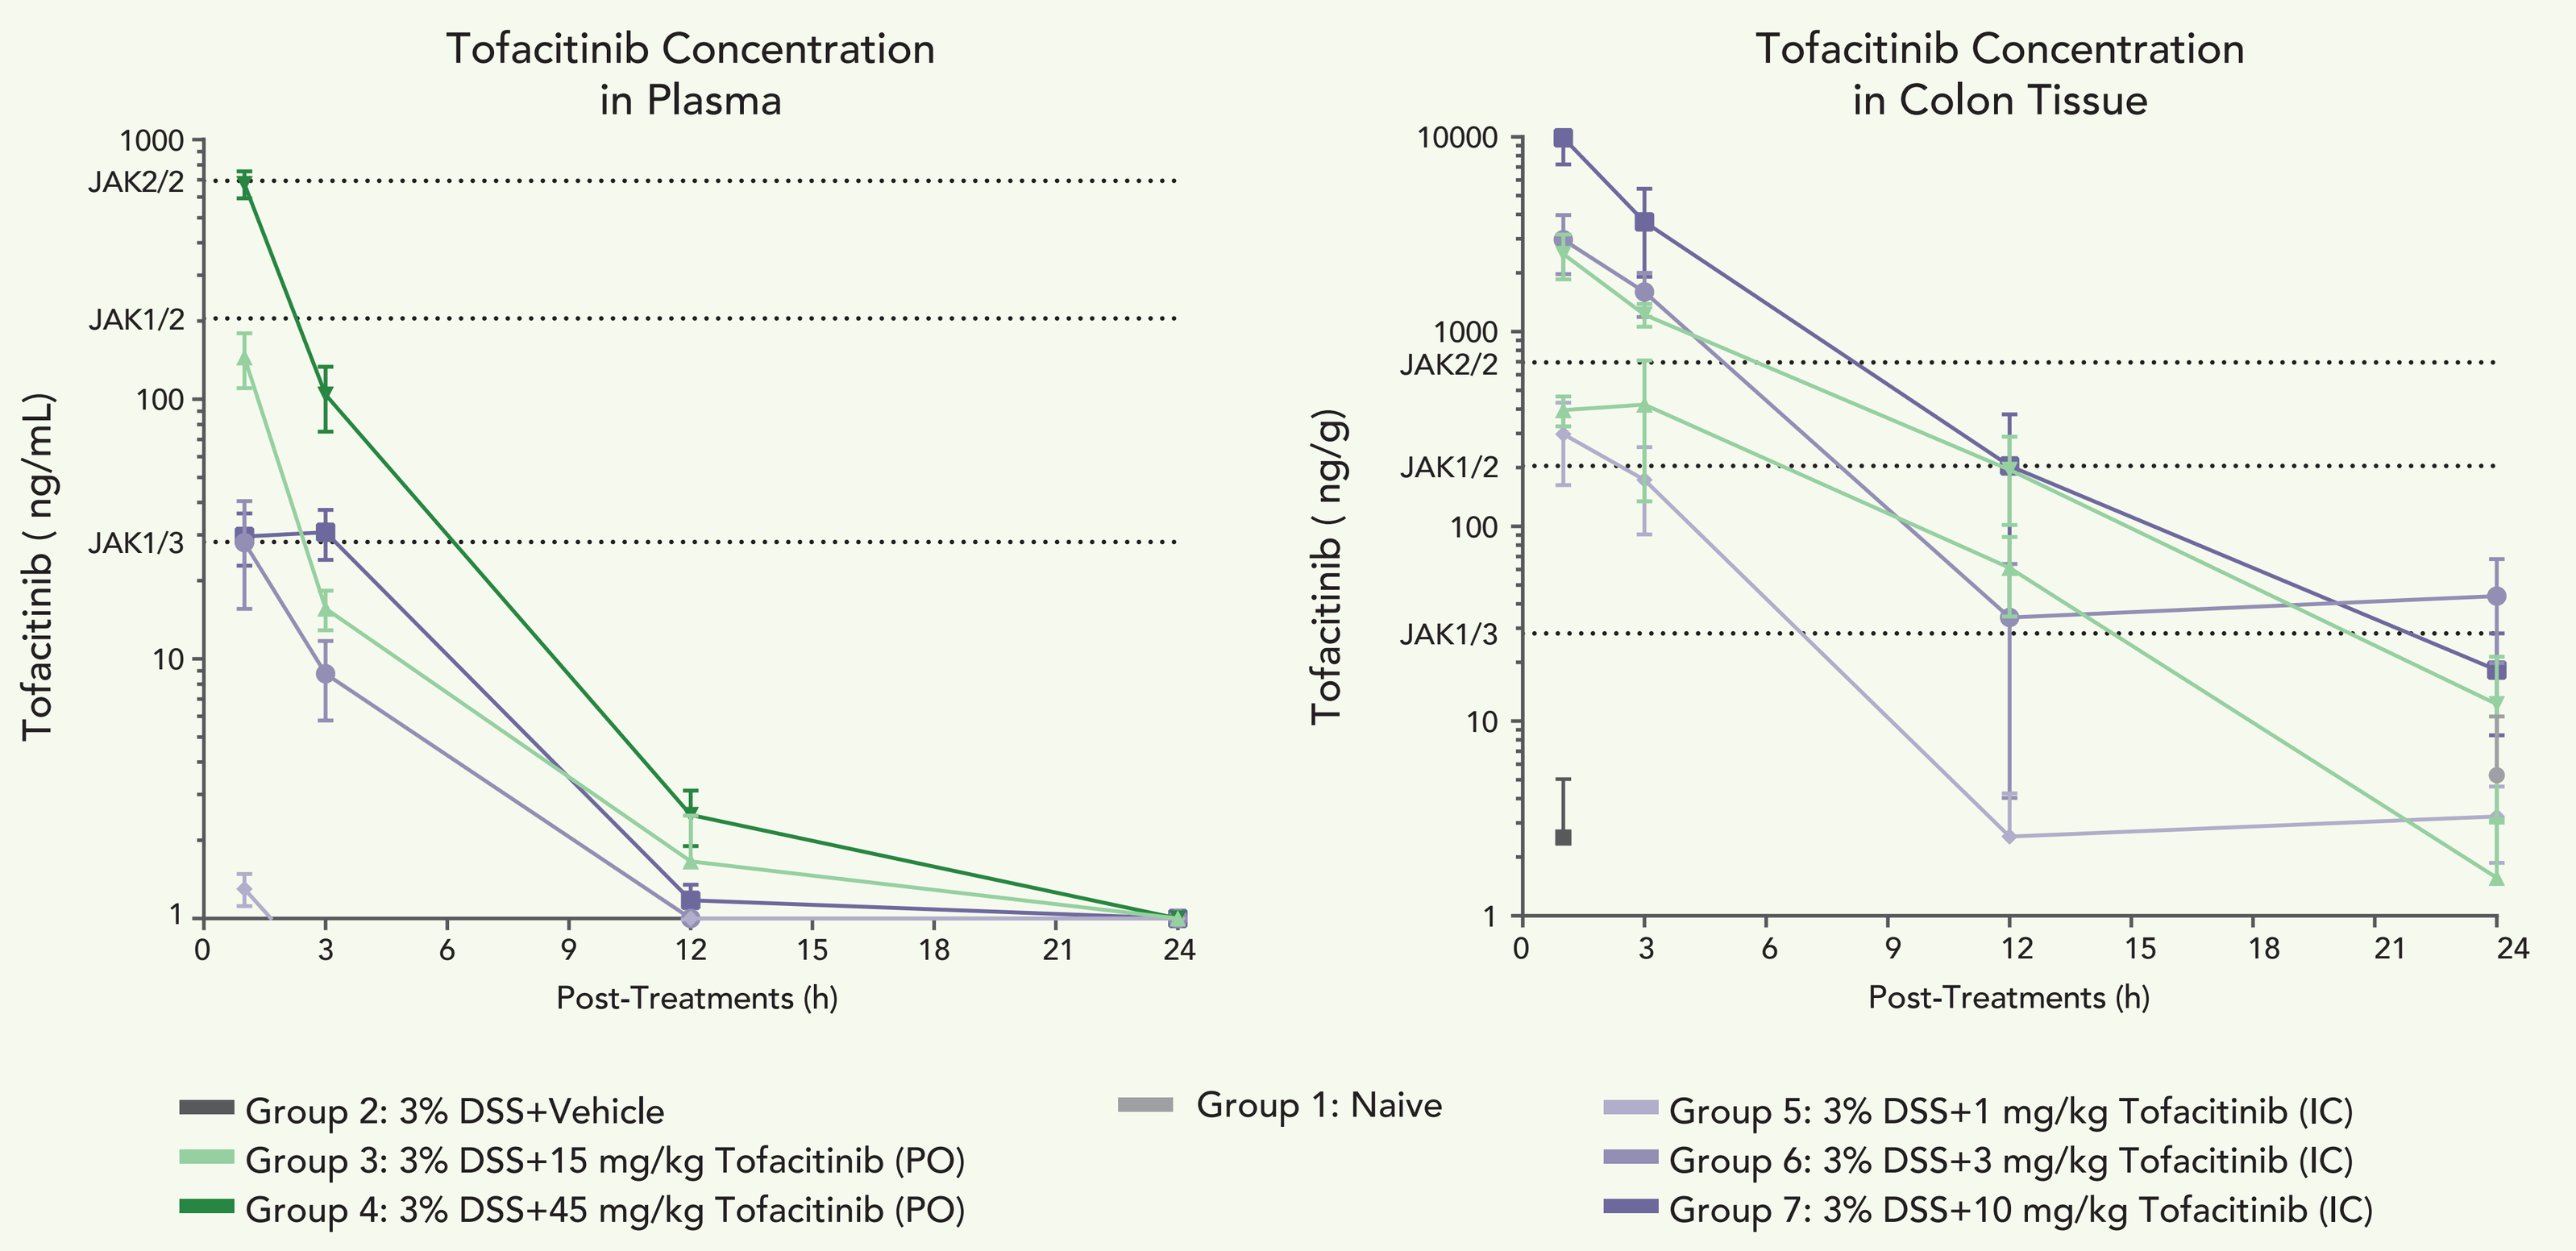
Tofacitinib Concentration in Plasma
Tofacitinib (ng/mL)
JAK2/2
JAK1/2
JAK1/3
Post-Treatments (h)
Tofacitinib Concentration in Colon Tissue
Tofacitinib (ng/g)
Group 2: 3% DSS+Vehicle
Group 3: 3% DSS+15 mg/kg Tofacitinib (PO)
Group 4: 3% DSS+45 mg/kg Tofacitinib (PO)
Group 1: Naive
Group 5: 3% DSS+1 mg/kg Tofacitinib (IC)
Group 6: 3% DSS+3 mg/kg Tofacitinib (IC)
Group 7: 3% DSS+10 mg/kg Tofacitinib (IC)

Figure 1. Mean plasma (left) and colon tissue (right) tofacitinib concentrations over 24 hours post-dose. Dashed line indicates *in vitro* IC_{50} of JAK1/3, JAK1/2, and JAK2/2 in whole blood.[1] Error bars represent standard deviation.

- Limited systemic exposure and equivalent tissue exposure were observed with a 10- to 15-fold lower IC once-daily (QD) dose than its corresponding PO groups (**Figure 2A**).
- PK modeling of Tofa colon tissue concentration with reported IC_{50} of JAKs[1] showed better effects for % of JAKs inhibtion over time via IC at 10–15X lower PO dose (**Figure 2B**).

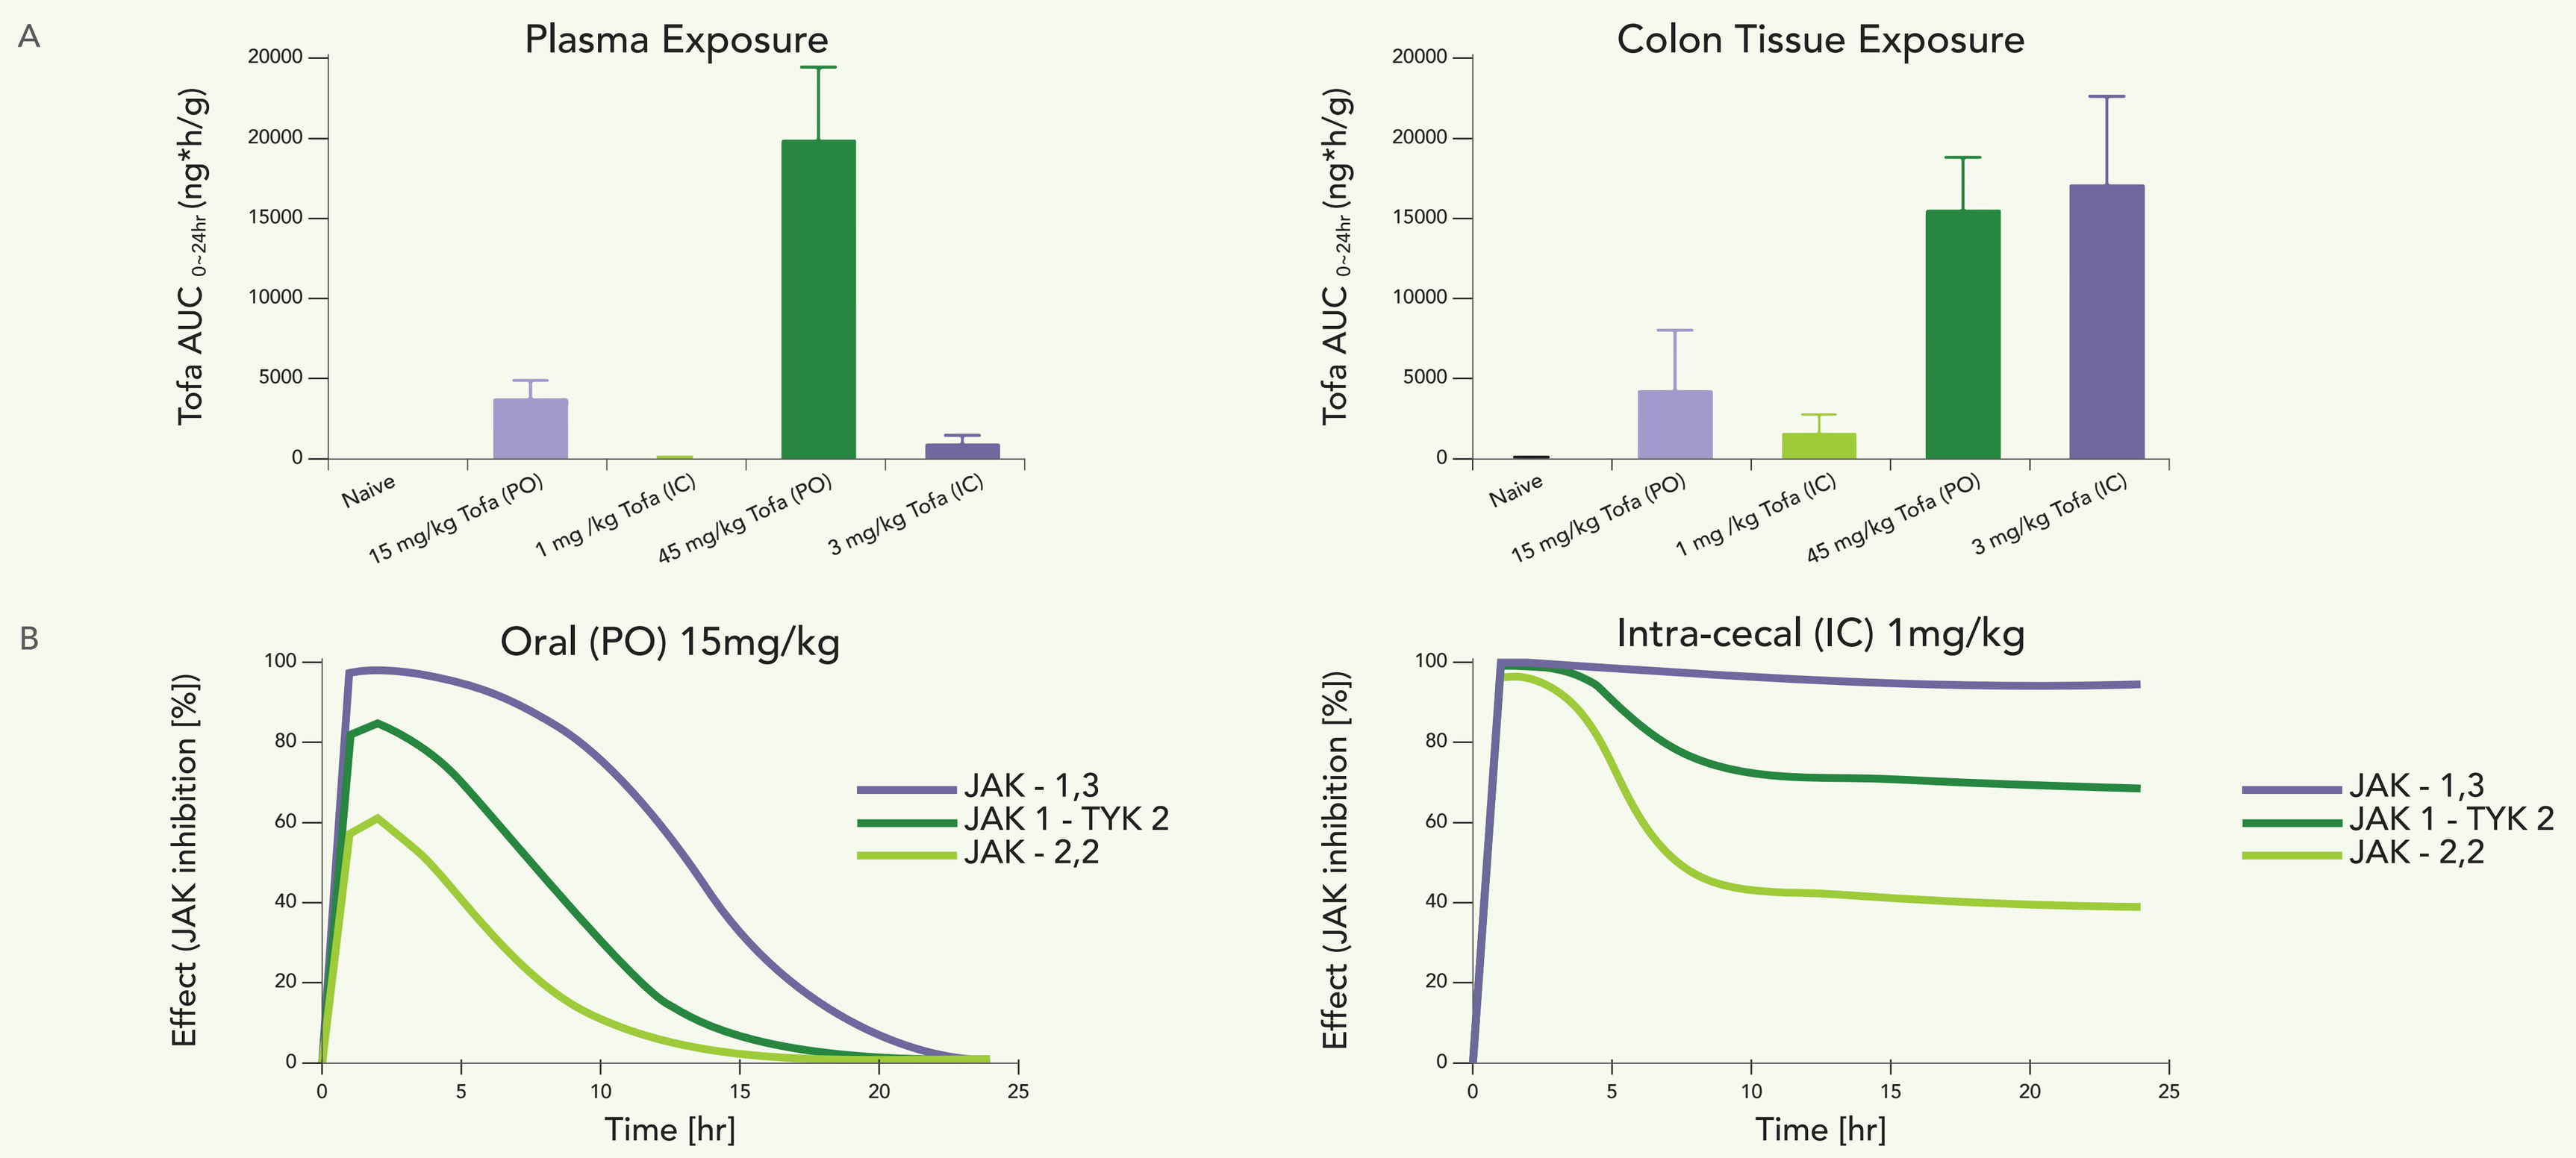
A
Plasma Exposure
Colon Tissue Exposure
Tofa AUC 0~24hr (ng*h/g)
Naive
15 mg/kg Tofa (PO)
1 mg /kg Tofa (IC)
45 mg/kg Tofa (PO)
3 mg/kg Tofa (IC)
B
Oral (PO) 15mg/kg
Intra-cecal (IC) 1mg/kg
Effect (JAK inhibition [%])
Time [hr]
JAK - 1,3
JAK 1 - TYK 2
JAK - 2,2

Figure 2. Pharmacokinetics of tofacitinib through oral (PO) vs. intra-cecal (IC) administration. (A) Plasma exposure (left panel) and colon tissue exposure (right panel) of Tofa given PO vs IC in DSS-colitis C57BL/6 mice at 0–24 hours. (B) Estimated JAK inhibition using PK modeling with tofacitinib concentration over time and reported IC_{50} of JAKs.[1]

Intra-cecal delivery of Tofa to the inflamed mucosa can potentiate PD effects in tissue at a lower treatment dose.

- The levels of pSTAT3 were most significantly induced in the control group, suggesting DSS-colitis may be mediated by the pSTAT3/IL-6 signaling pathways (**Figure 3**).
- IC QD Tofa at 10- to 15- fold lower doses showed greater efficacy at inhibiting pSTAT3 in the colon over time and compared to higher oral doses (**Figure 3**).

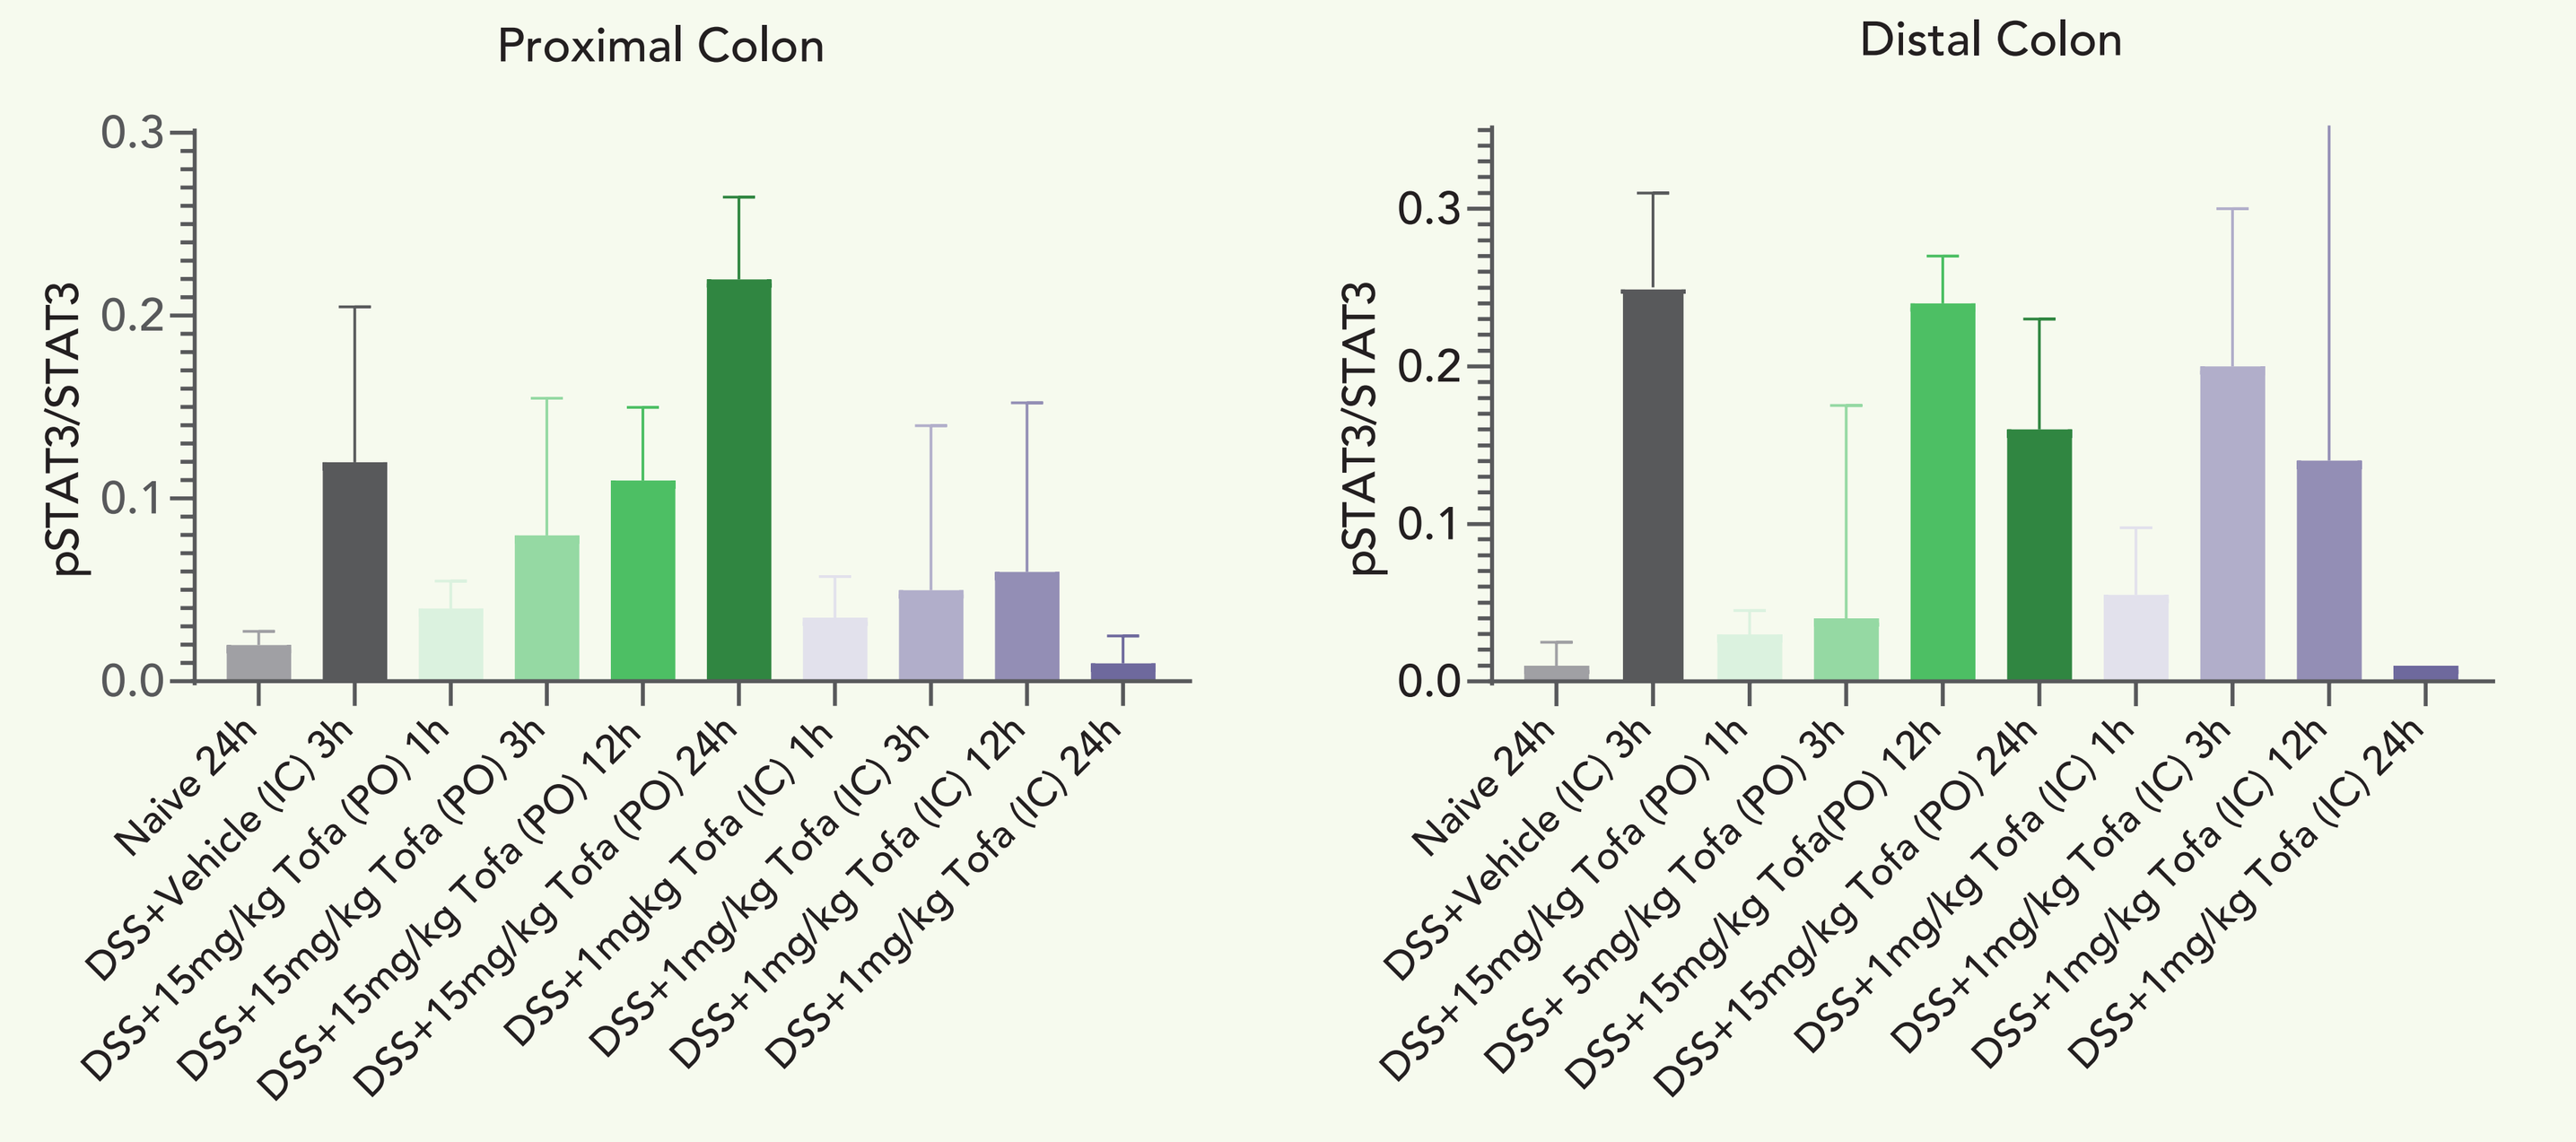
Proximal Colon
Distal Colon
pSTAT3/STAT3
Naive 24h
DSS+Vehicle (IC) 3h
DSS+15mg/kg Tofa (PO) 1h
DSS+15mg/kg Tofa (PO) 3h
DSS+15mg/kg Tofa (PO) 12h
DSS+15mg/kg Tofa (PO) 24h
DSS+1mgkg Tofa (IC) 1h
DSS+1mg/kg Tofa (IC) 3h
DSS+1mg/kg Tofa (IC) 12h
DSS+1mg/kg Tofa (IC) 24h
DSS+ 5mg/kg Tofa (PO) 3h
DSS+15mg/kg Tofa(PO) 12h

Figure 3. pSTAT3/STAT3 ratio in proximal and distal colon tissue. Median pSTAT3/STAT3 Positive Area %; ±SEM).

Soluble Tofa formulation increased tissue absorption and coverage via IC administration.

- A dose-dependent increase in tissue absorption and coverage in the colon following four repeated IC QD Tofa soluble formulation (TSF) was observed compared to oral suspension formulation (TOF) (**Figure 4**).
- Follow-up formulation evaluation studies in IC swine also demonstrated that improved tissue absorption and coverage in the distal GI tract could be achieved with improved solubility (data not shown).

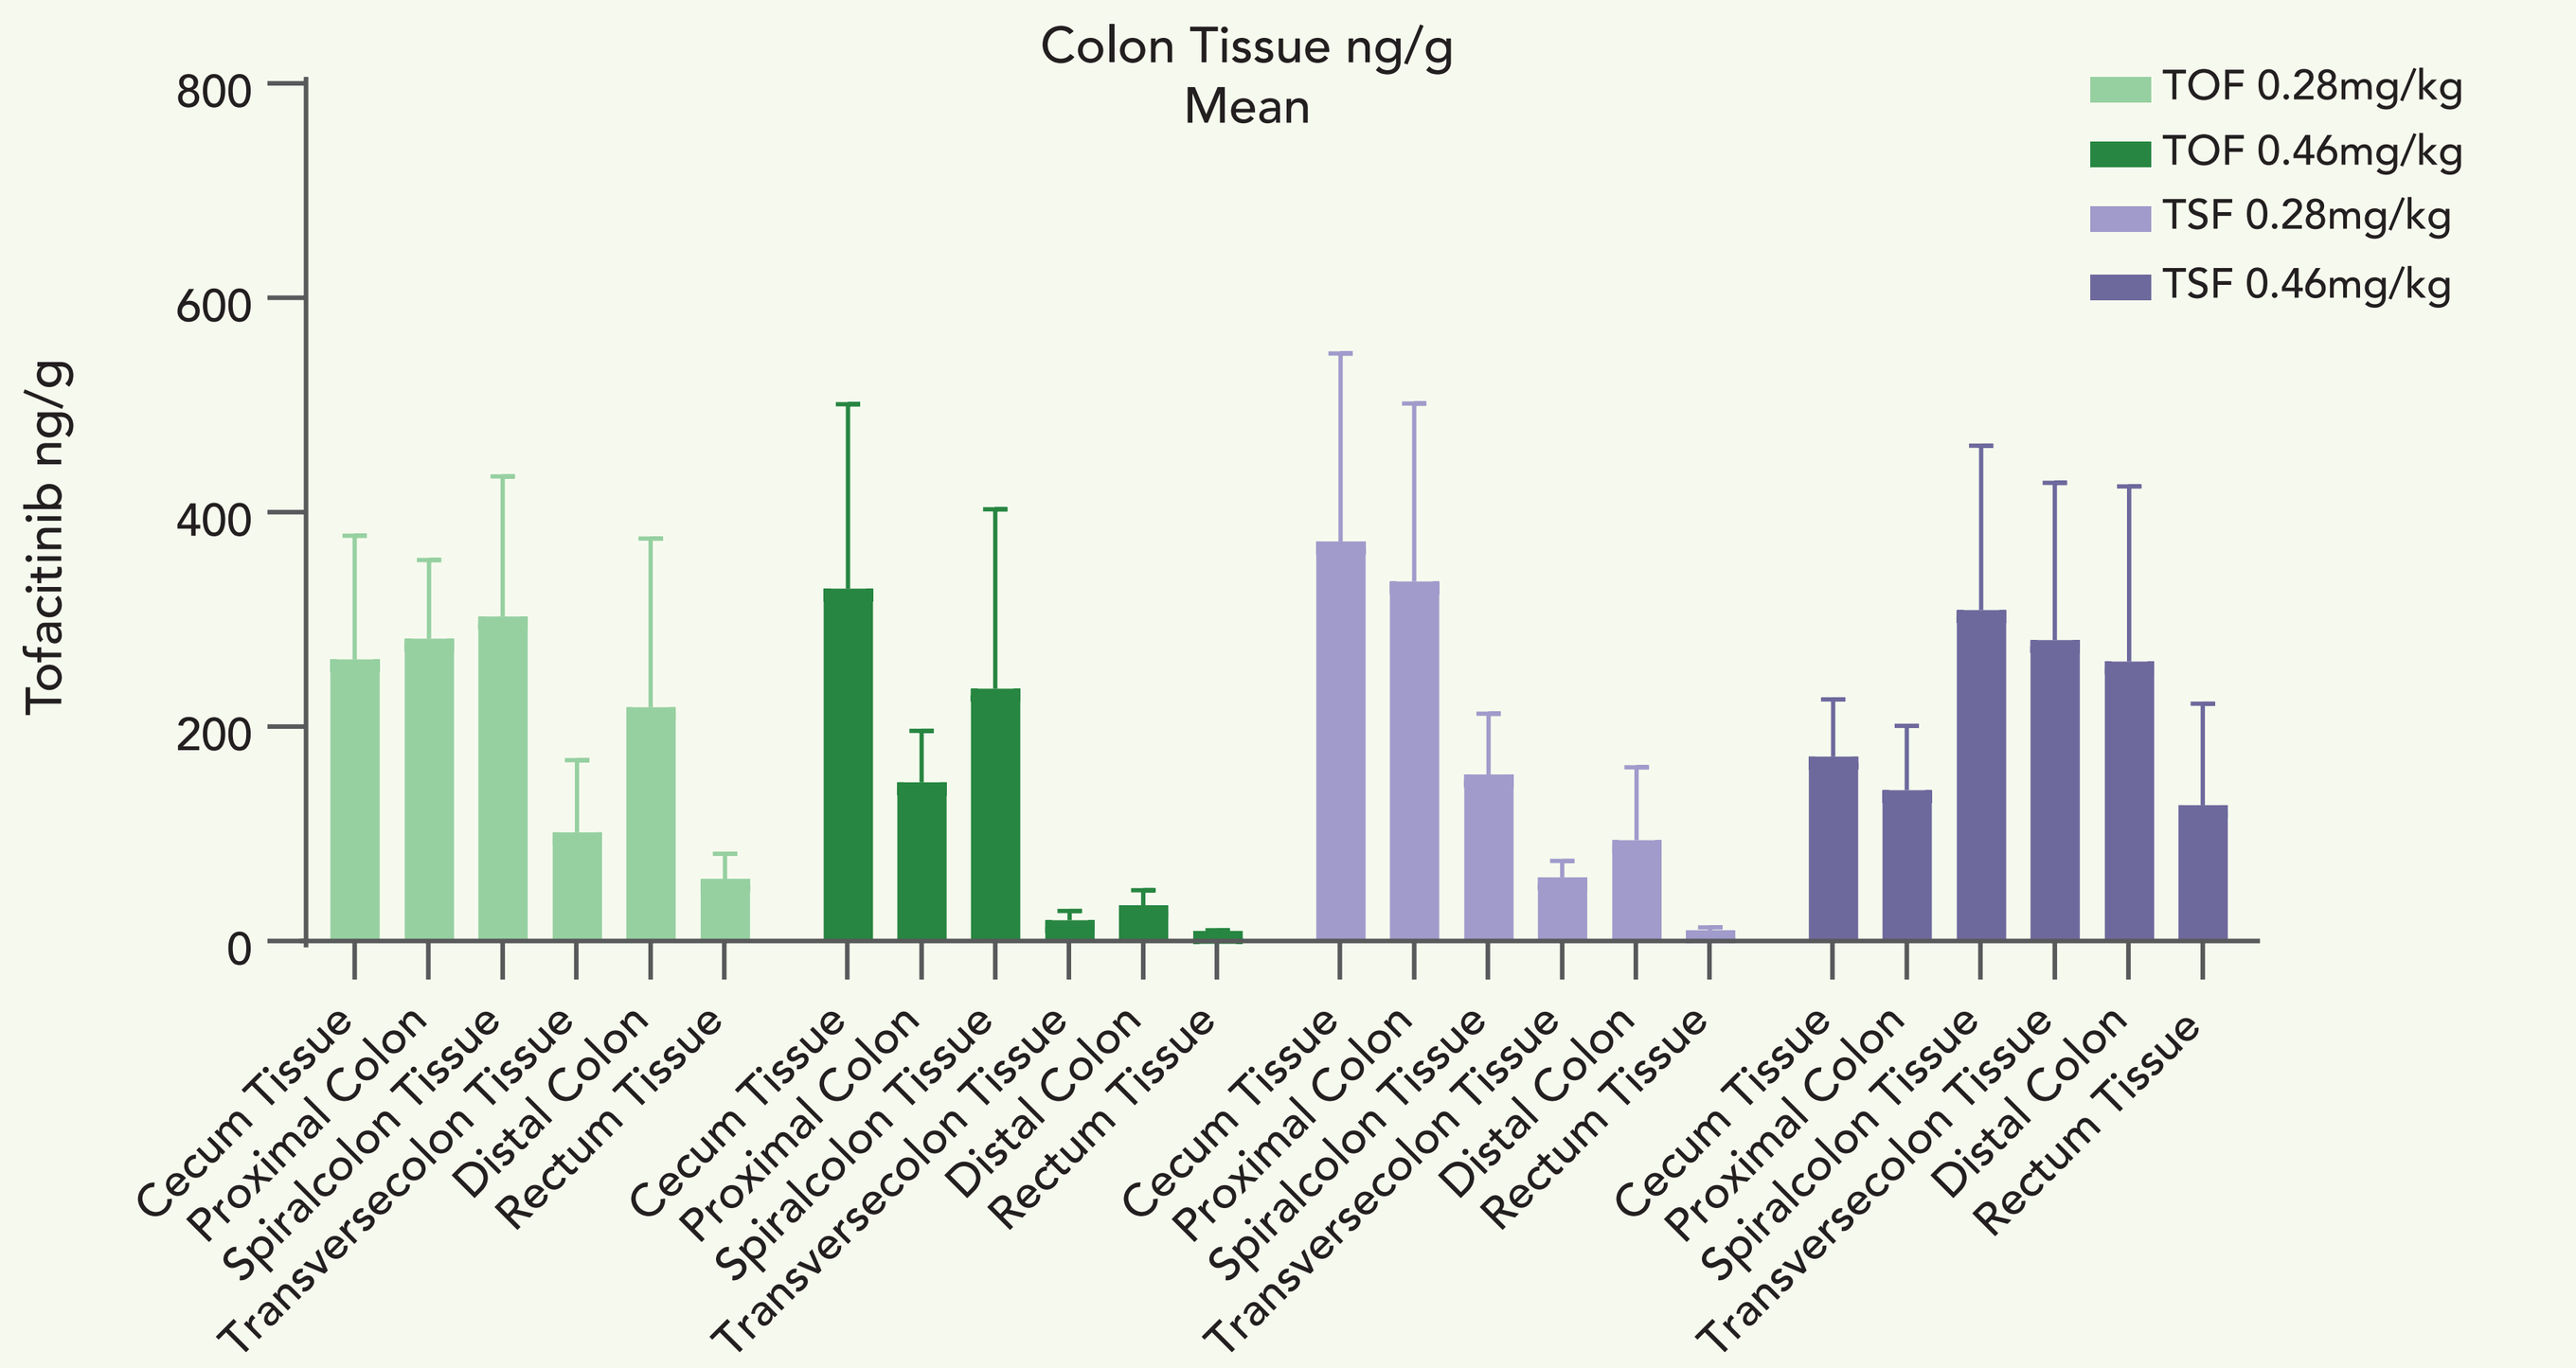
Colon Tissue ng/g
Mean
Tofacitinib ng/g
TOF 0.28mg/kg
TOF 0.46mg/kg
TSF 0.28mg/kg
TSF 0.46mg/kg
Cecum Tissue
Proximal Colon
Spiralcolon Tissue
Transversecolon Tissue
Distal Colon
Rectum Tissue

Figure 4. Mean tofacitinib tissue concentrations in cecum, proximal to distal colon, and rectum tissues in swine (ng/g) following four repeated intra-cecal once-daily administrations. Different colors represent two different dose concentrations of tofacitinib citrate, 0.28mg/kg (light green and light purple) and 0.46 mg/kg (dark green and dark purple). Error bars represent standard deviation. TOS = Tofacitinib Oral Suspension; TSF = Tofacitinib Soluble Formulation.

Conclusion

These results indicate that the targeted delivery of solubilized tofacitinib to the site of inflammation could increase tissue absorption and coverage to achieve maximum efficacy with a lower risk of systemic toxicity.

References

1. Meyer D, Jesson M, Li X, et al. Anti-inflammatory activity and neutrophil reductions mediated by the JAK1/JAK3 inhibitor, CP-690,550, in rat adjuvant-induced arthritis. *J Inflamm* 2010; 7: 41.
2. Neubauer H, Cumano A, Muller M, et al. Jak2 deficiency defines an essential developmental checkpoint in definitive hematopoiesis. *Cell* 1998; 93(3): 397-409.
3. Egea L, Hirata Y, Kagnoff M. GM-CSF: a role in immune and inflammatory reactions in the intestine. *Expert Rev Gastroenterol Hepatol* 2010; 4(6): 723 31.
4. Beattie D, Pulido-Rios M, Shen F, et al. Intestinally-restricted Janus Kinase inhibition: a potential approach to maximize the therapeutic index in inflammatory bowel disease therapy. *J Inflamm* 2017; 14: 28.

Contact

Shaoying Nikki Lee
Progenity Inc.

4330 La Jolla Village Drive, Suite 200
San Diego, CA 92121

Nikki.Lee@progenity.com

©2021 Progenity, Inc. All rights reserved.
Progenity® is a registered service mark of Progenity, Inc. | **progenity.com/research**