Tofacitinib tissue exposure correlates with endoscopic outcome

Bram Verstockt[1,2], Dahham Alsoud[2], Joep van Oostrom[3], Jeff Smith[4], Jack Styli[4], Sujay Singh[4], Sarah van Gennep[3], Pejman Rahimian[4], João Sabino[2,3], Marc Ferrante[1,2], Sharat Singh[4], Geert D'Haens[3], Séverine Vermeire[1,2]

1.Department of Gastroenterology and Hepatology, University Hospitals Leuven, KU Leuven, Leuven, Belgium 2. Translational Research in Gastrointestinal Disorders, Department of Chronic Disease and Metabolism, KU Leuven, Leuven, Belgium 3.Department of Gastroenterology and Hepatology, Amsterdam UMC, Amsterdam, The Netherlands. 4. Progenity Inc, San Diego California, USA.

www.ibd-leuven.com

bram.verstockt@uzleuven.be

@bverstockt
@ibdleuven

KU LEUVEN

P401

BACKGROUND

- **Tofacitinib** is the **1st in class** Janus Kinase (JAK) inhibitor, approved for moderate-to-severely active **ulcerative colitis** (UC).
- In contrast to monoclonal antibodies, **very little** is **known** about **pharmacokinetic-pharmacodynamic profile** of **small molecules**.

AIM:

- To assess **pharmacokinetic-pharmacodynamic changes** in **tofacitinib** (TFC) treated UC patients
- To assess the **potential of STAT3 phosphorylation as predictive / monitoring tool**, as it has been proposed as a marker of efficacy mechanistically

METHODS

- **30 consecutive UC patients** with **active endoscopic disease** (Mayo endoscopic subscore 2-3) **initiating TFC** were recruited at the University Hospitals Leuven (Belgium) and the Amsterdam UMC (The Netherlands) (Table 1)

Sex, women, n (%)	17 (56.7)
Age at initiation of tofacitinib, y, median [IQR]	47.1 (28.2 – 53.7)
Disease duration, y, median [IQR]	7.5 (3.1 – 15.4)
Faecal calprotectin, mg/g, median [IQR]	1718.6 (1446.8 – 1800)
Serum albumin, g/L, median [IQR]	41.7 (40.8 – 44.2)
Partial Mayo score, median [IQR]	10.0 (9.0 – 11.0)
Endoscopic scoring	
- **Mayo endoscopic sub-score, median [IQR]**	3.0 (2.0 – 3.0)
- **Ulcerative colitis index of severity, median [IQR]**	5.0 (4.0 – 6.0)
Histologic scoring	
- **Nancy index, median [IQR]**	3.0 (3.0 – 3.0)
Disease extent, n (%)	
- **Proctitis E1**	4 (13.3)
- **Left-sided colitis E2**	14 (40.0)
- **Extensive colitis E3**	12 (46.7)
Concomitant medication, n (%)	
- **Corticosteroids**	
• **Topical**	7 (23.3)
• **Systemic**	8 (26.7)
- **Immunomodulators**	0 (0.0)
Smoking status, n (%)	
- **Active smoking**	4 (13.3)
- **Previously smoking**	10 (33.3)
- **Never smoked**	16 (53.3)

Table 1: Baseline features

- Patients **initiated TFC 10mg BID till week 8**, with subsequent **de-escalation to 5mg BID upon discretion of the treating physician**

- Patients were **prospectively** monitored, with **sampling** (colonic tissue and serum) at **baseline and 8-16 weeks after TFC initiation** (Figure 1).

- **Endoscopic improvement** was defined as a **Mayo endoscopic subscore 0-1**.

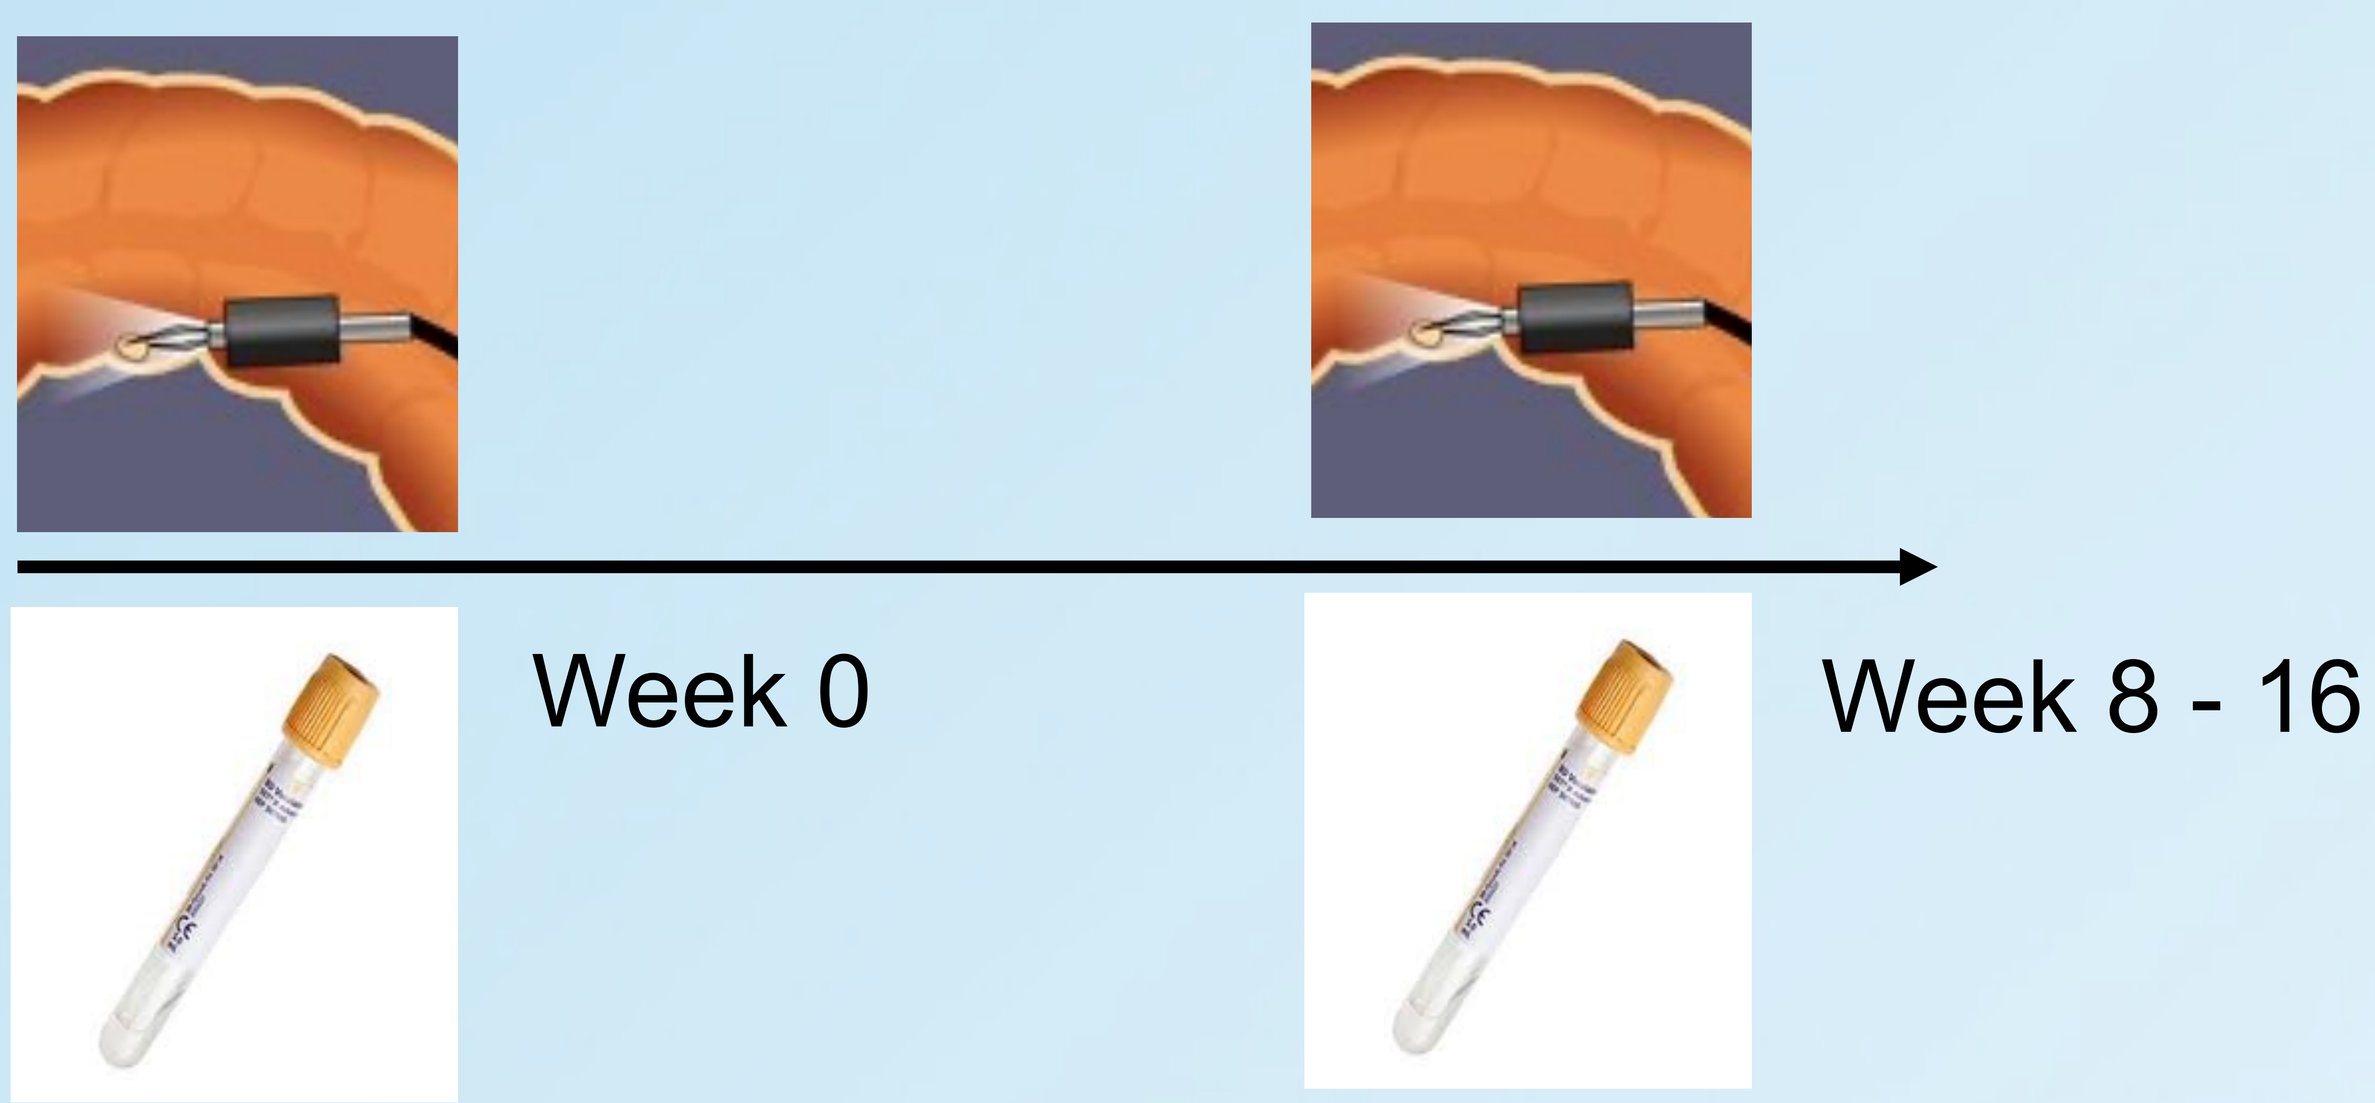
Week 0
Week 8 - 16

Figure 1: Sampling strategy and prospective monitoring

- **TFC** was **extracted** from **tissue** using **acetonitrile, dried down** and **quantitated** using **mass spectrometry**. Both **total as well as phosphorylated** STAT3 were **measured in lysed tissue using specific antibodies** with an **ultrasensitive luminescent oxygen channelling assay**.

RESULTS

- **TFC tissue and serum concentrations correlated significantly** (r=0.92, p<0.001), though were **significantly higher in tissue** (median 520.19ng/g vs 17.35ng/mL, p<0.001) (Figure 2).

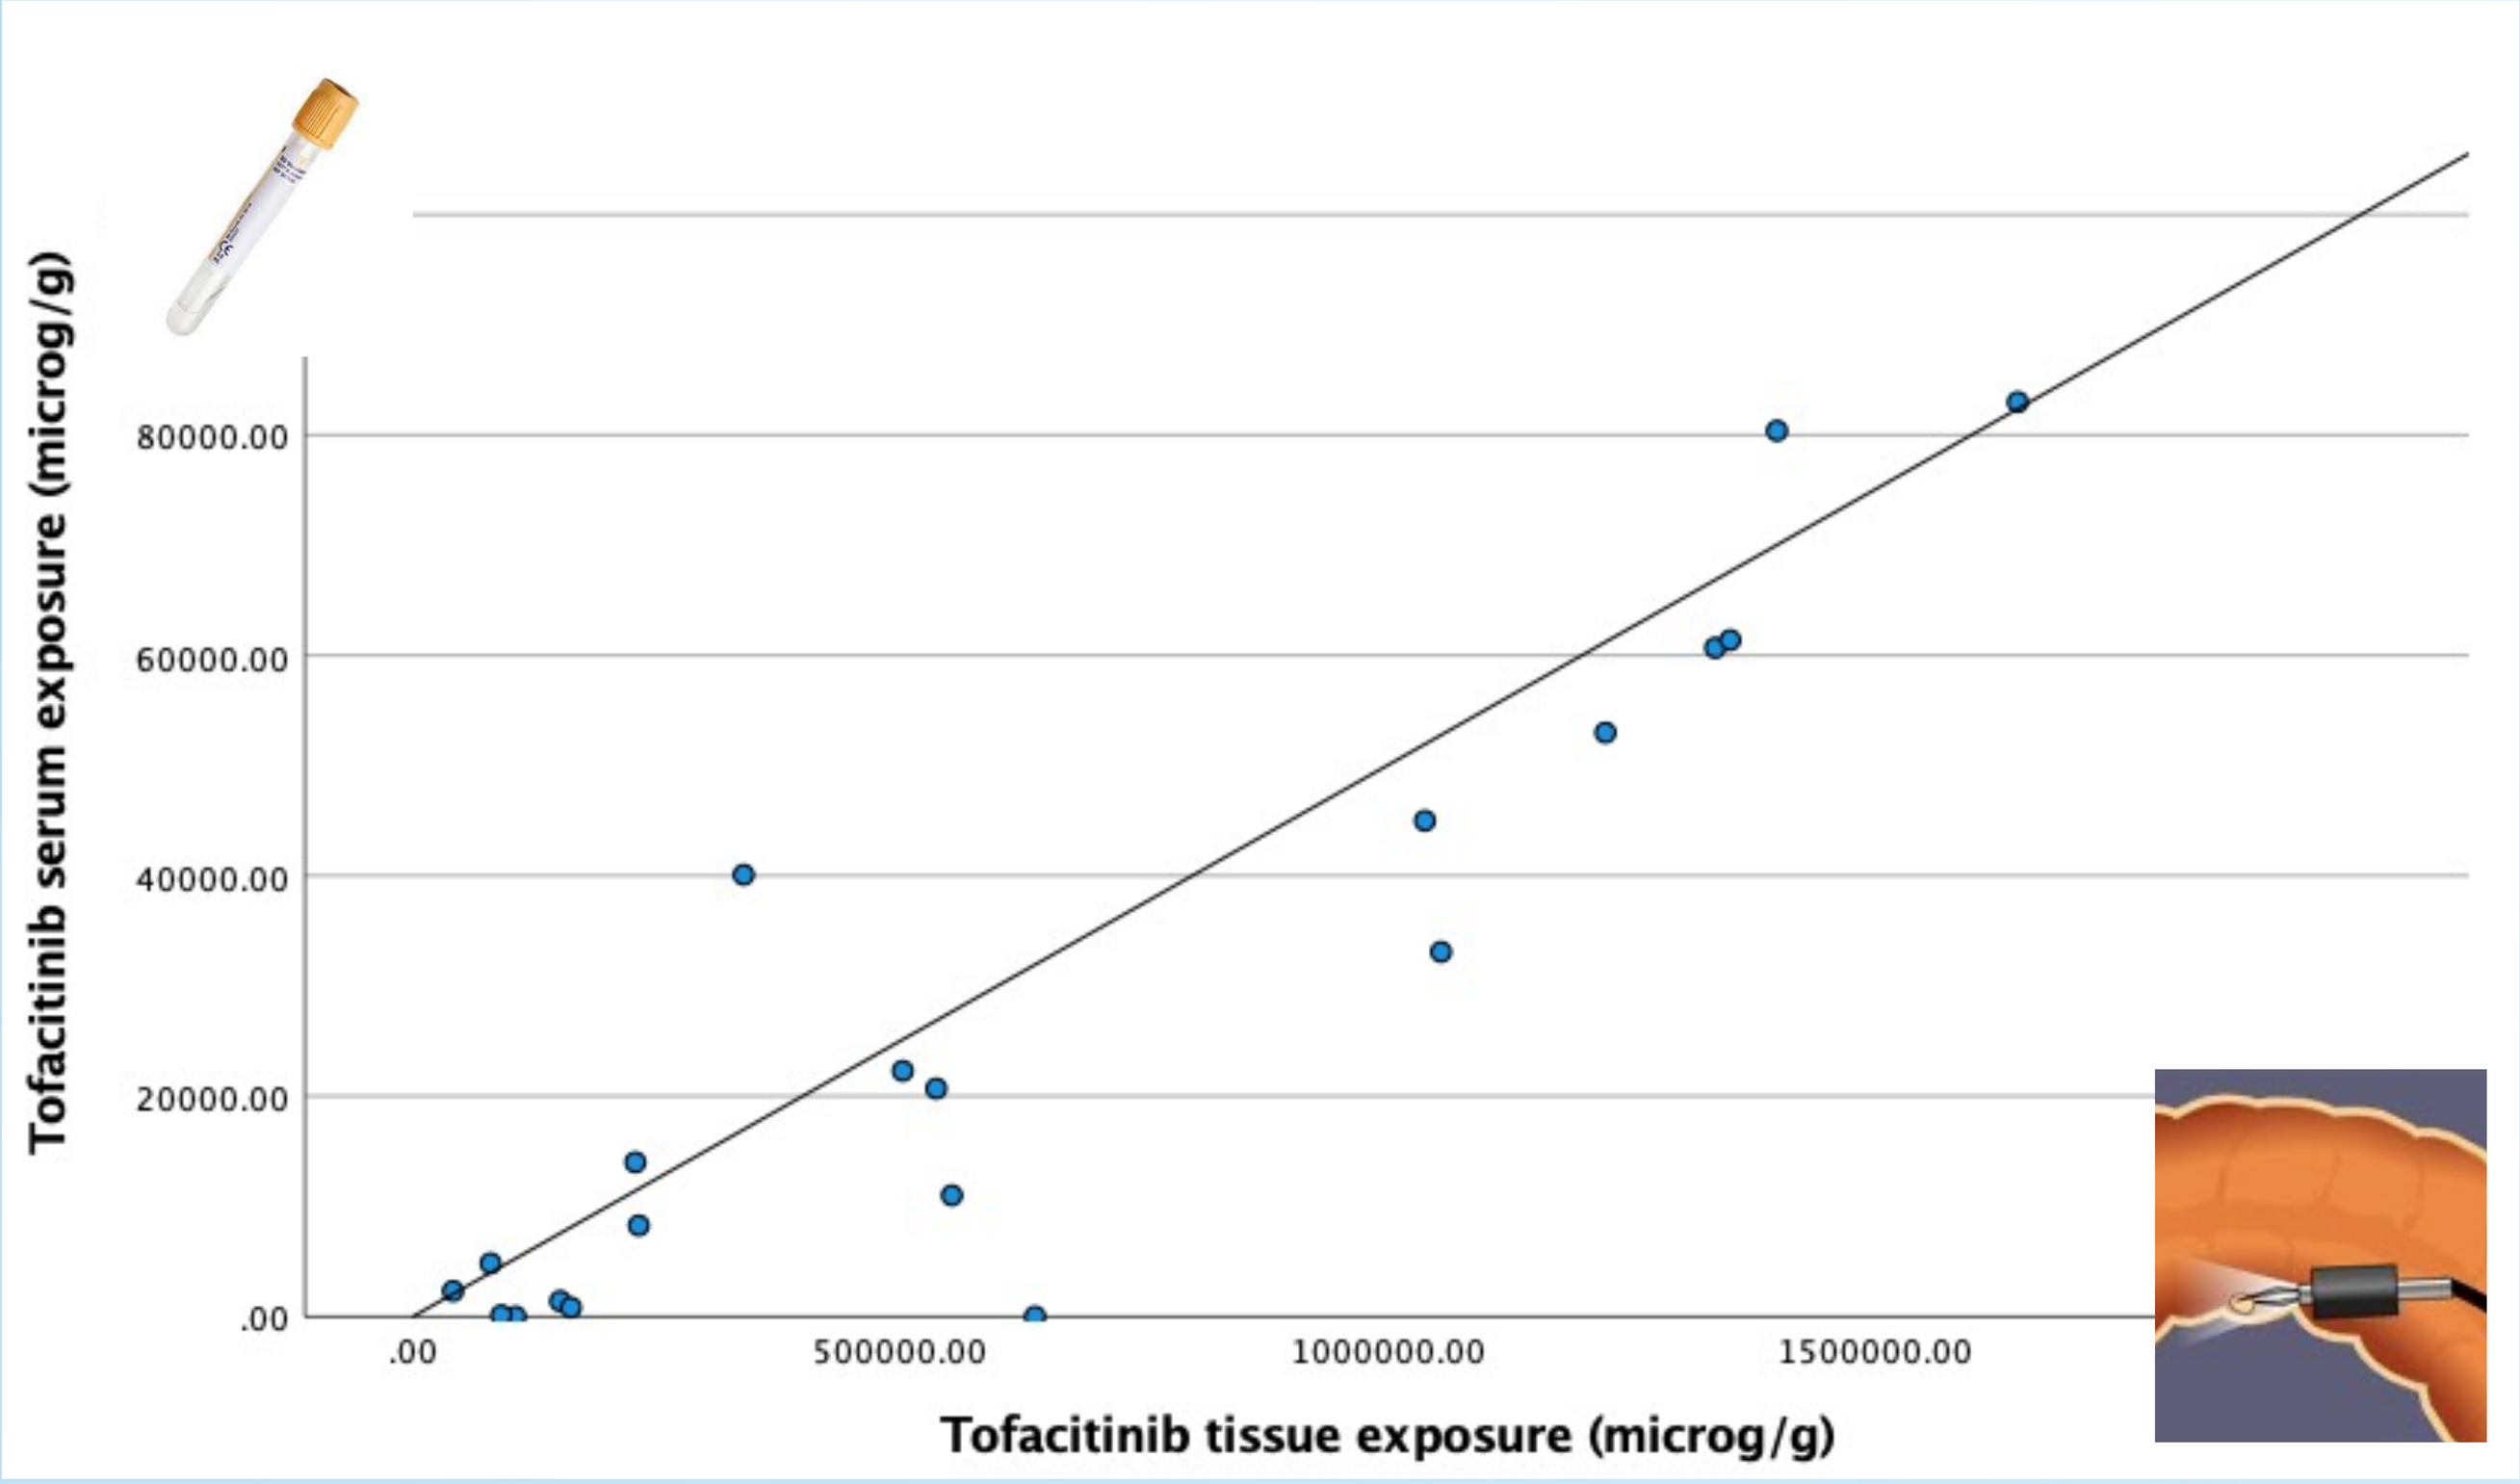
Tofacitinib serum exposure (microg/g)
80000.00
60000.00
40000.00
20000.00
.00
.00
500000.00
1000000.00
1500000.00
Tofacitinib tissue exposure (microg/g)

Figure 2: Correlation between TFC tissue and serum concentration

- In contrast to TFC serum exposure (p=0.26), **TFC tissue exposure** at the **end of induction was associated with endoscopic improvement by week 16** (p=0.04) (Figure 3).

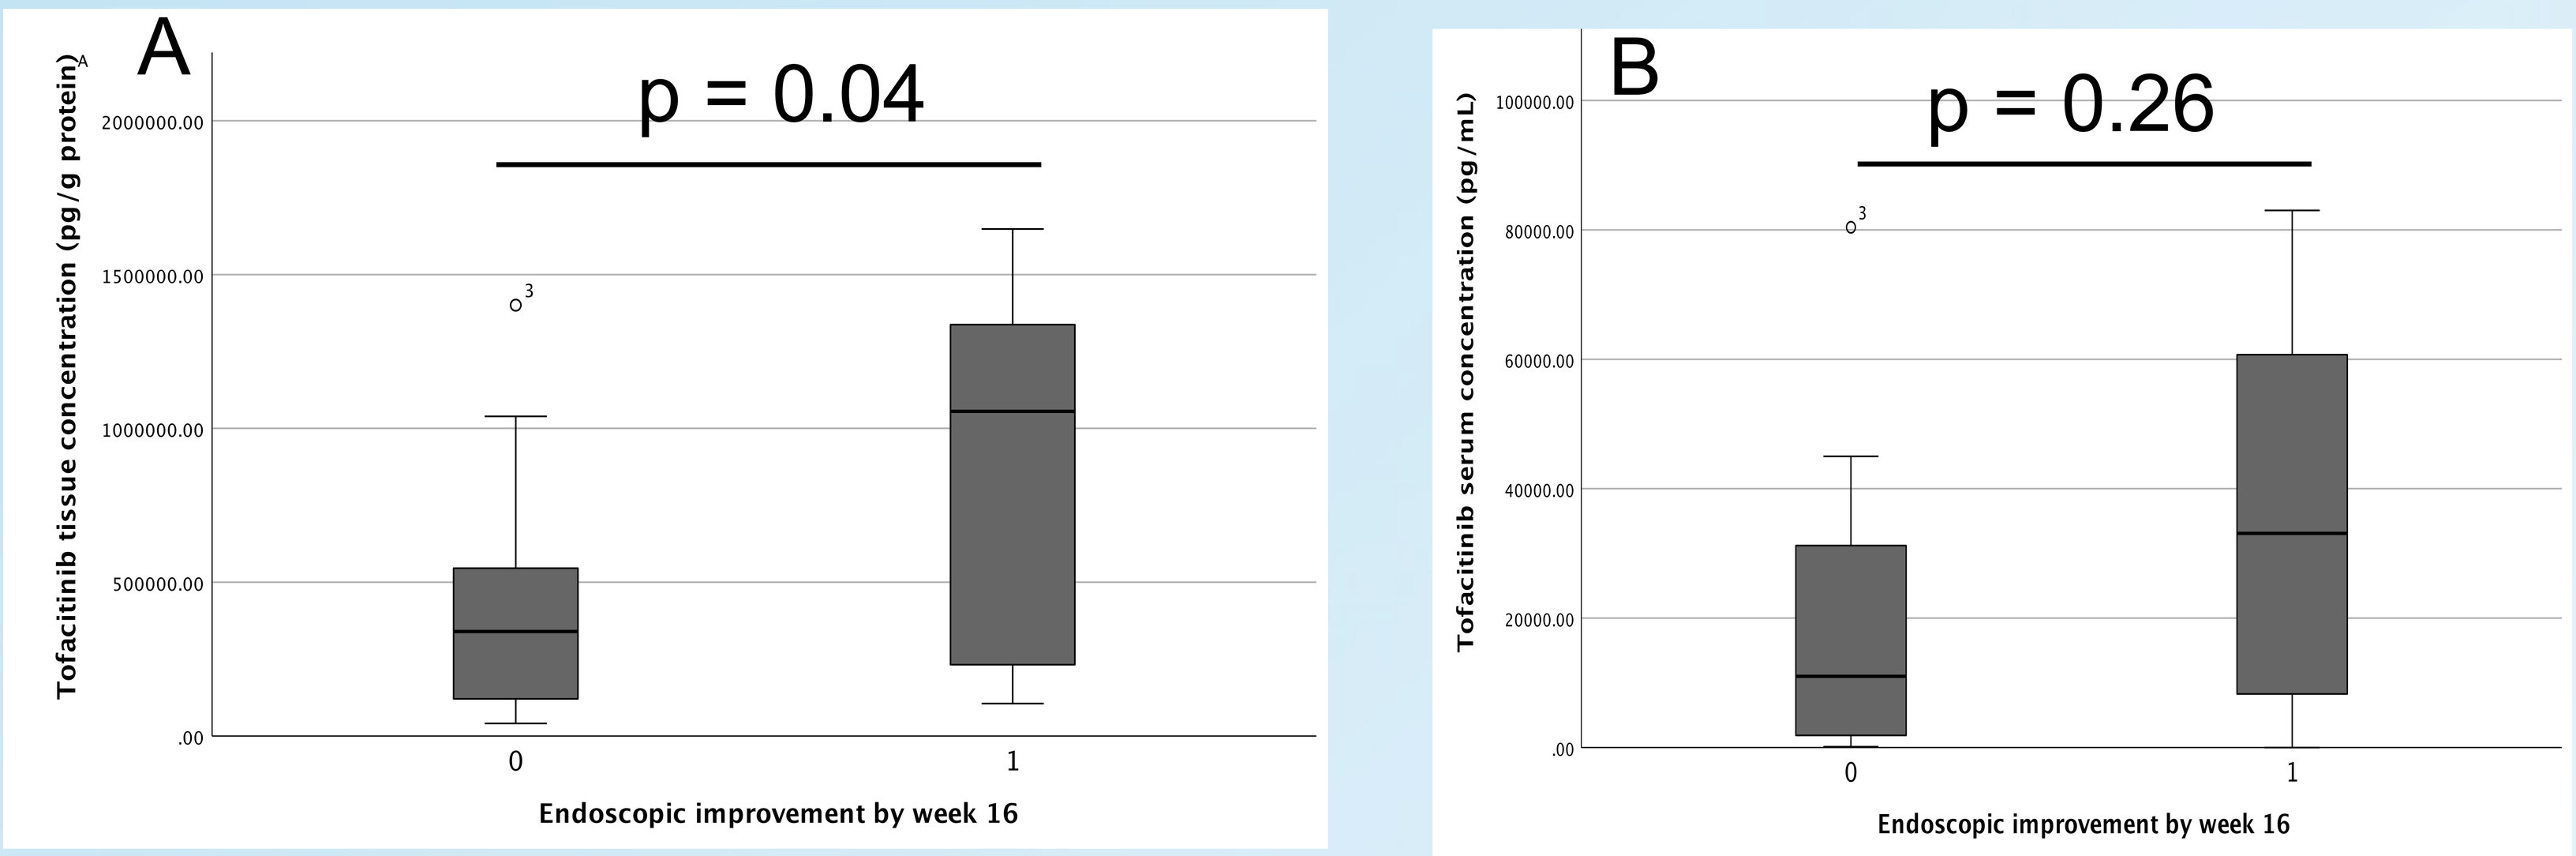
A
p = 0.04
Tofacitinib tissue concentration (pg/g protein)
Endoscopic improvement by week 16
B
p = 0.26
Tofacitinib serum concentration (pg/mL)
Endoscopic improvement by week 16

Figure 3: Comparison in tofacitinib tissue (A) or serum (B) exposure between (non-)responders by week 16

- In **TFC responders** (n=14), **TFC tissue exposure exceeded the concentration required to block 90% of the target (IC90)** reported in literature (median tissue exposure 1,055.00ng/g; IC90 823ng/g). **TFC tissue exposure in non-responders (n=16) was lower, but clearly exceeded the IC50**.

- Although **IL-6** was **not significantly downregulated** after TFC induction, a **significant decrease in the ratio of mucosal IL-6 driven phosphorylated STAT3 over total STAT3 (pSTAT3/STAT3) was observed in responders** (p=0.05), but not in non-responders (p=0.88) (Figure 4).

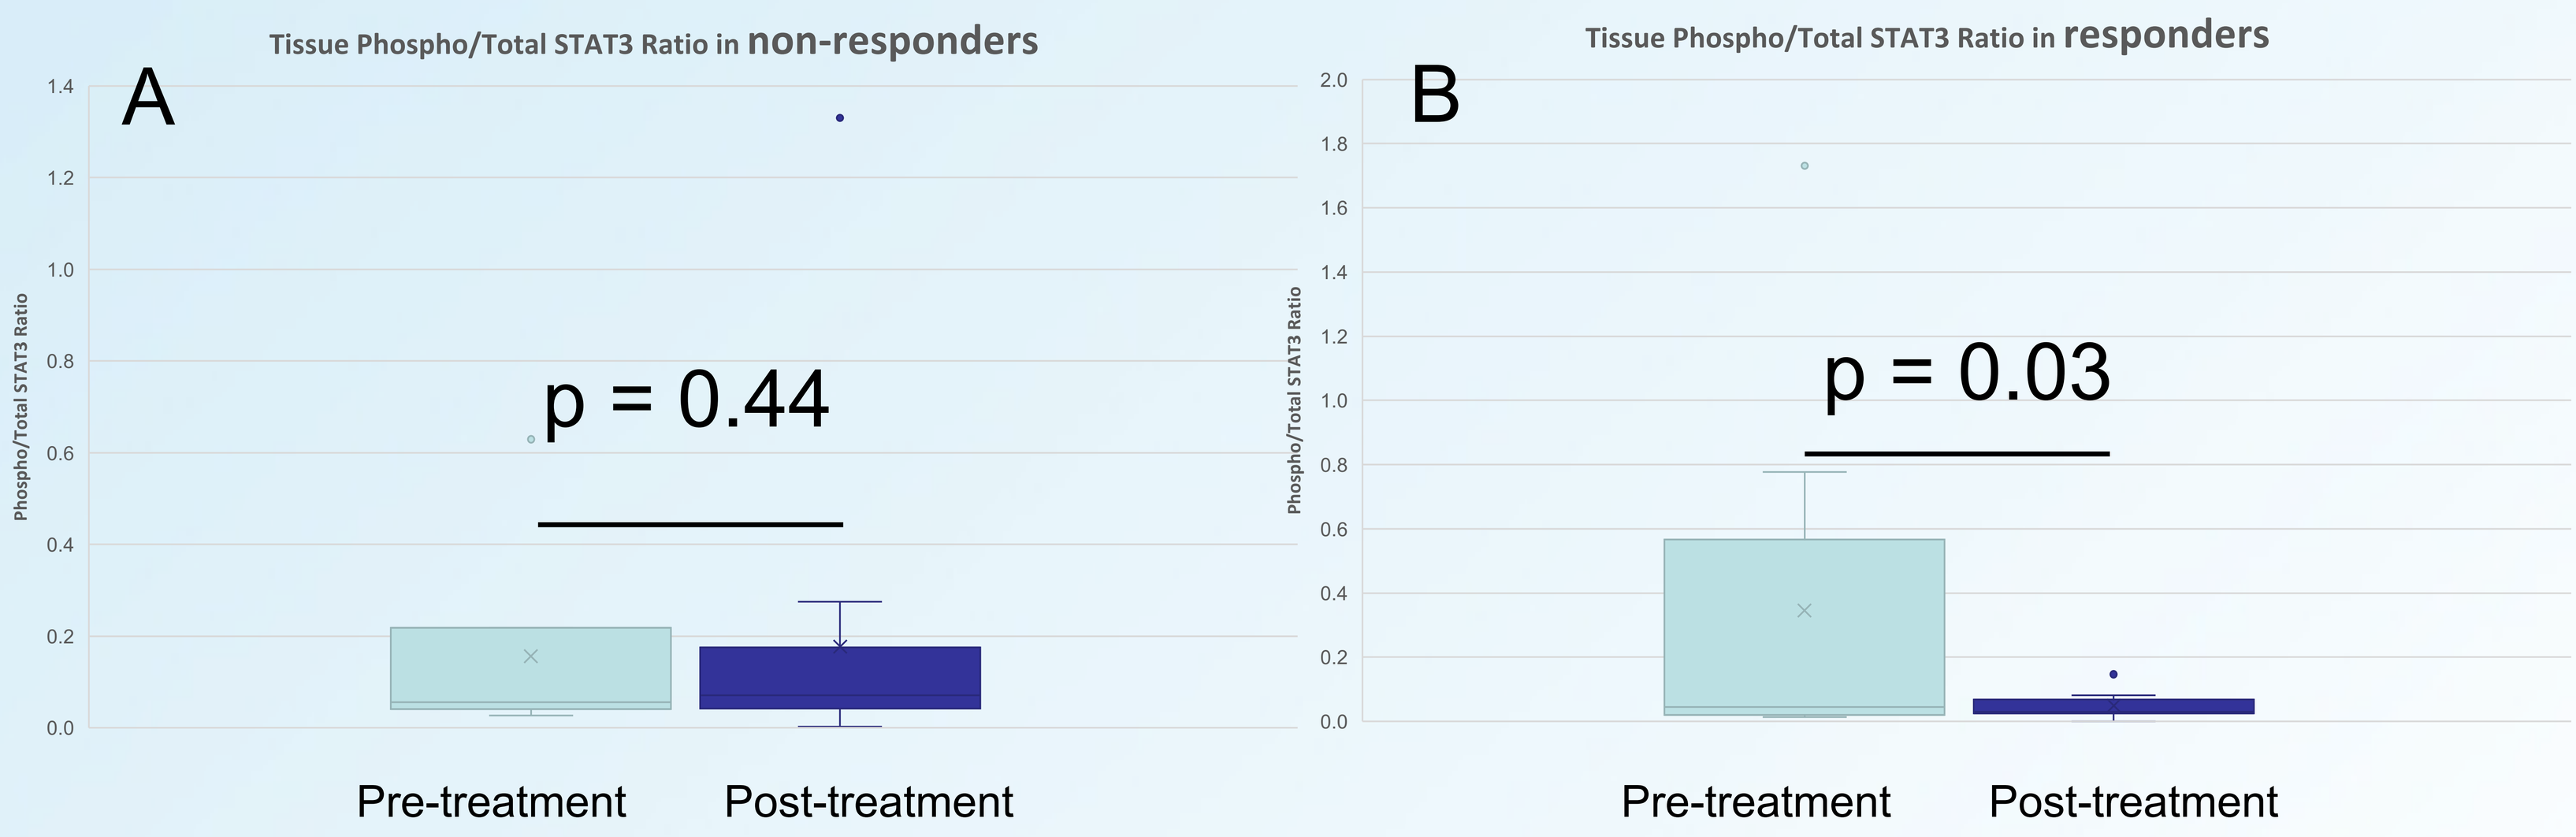
Tissue Phospho/Total STAT3 Ratio in non-responders
A
p = 0.44
Phospho/Total STAT3 Ratio
Pre-treatment
Post-treatment
Tissue Phospho/Total STAT3 Ratio in responders
B
p = 0.03
Phospho/Total STAT3 Ratio
Pre-treatment
Post-treatment

Figure 4: Tissue phospho/total STAT3 before and after TFC therapy in non-responders (A) and responders (B)

- The **pSTAT3/STAT3 ratio** also **correlated** significantly **with faecal calprotectin** (r=0.35, p=0.05), but **only weakly with the Mayo endoscopic sub score** (r=0.22, p=0.13).

- **Baseline** mucosal **pSTAT3/STAT3 did not differ significantly between future responders and non-responders**.

CONCLUSIONS

- We demonstrated for the first time **a mucosal exposure-response relationship** with tofacitinib in UC
- **pSTAT3/STAT3 ratio** is a potential **molecular marker to track response**, directly linked to the MOA of tofacitinib
- Can an increased local dose of tofacitinib result in better efficacy without compromising safety? **UC induction trial with PGN-600** (topical tofacitinib) soon to be **expected**.