Development of *ex-vivo* and *in-vivo* models to assess the performance of an oral biotherapeutic delivery system (OBDS) capsule

Shaoying Nikki Lee[1], Cheryl Stork[1], Jeff Smith[1], Bryan Smith[1], Nelson Quintana[1], Chris Wahl[1], Sharat Singh[1]
1 Biora Therapeutics Inc., CA, USA

BIORA™ Therapeutics

INTRODUCTION

Biologics/peptides/nucleic acids are highly effective drugs; however, oral delivery of these therapeutics has proven difficult due to the harsh conditions of the upper gastrointestinal tract (GIT) and poor absorption in the small intestinal mucosa. The current state of-the-art technology for a successful oral protein delivery provides around 1% bioavailability when delivered as an oral tablet (Rybelsus® oral Semaglutide). We aim to develop an oral biotherapeutic delivery system (OBDS) that prevents drug degradation in the upper GIT and increases bioavailability via submucosal injection.

THE OBDS DEVICE

- The autonomous OBDS device is comprised of a drug module, which houses a formulation of a therapeutic compound, and a trigger module under pressurized gas control **(Figure 1)**.
- The OBDS capsule operates autonomously and provides a needleless injection to deposit the liquid drug payload into the submucosal space of the proximal small intestine for absorption by systemic circulation.

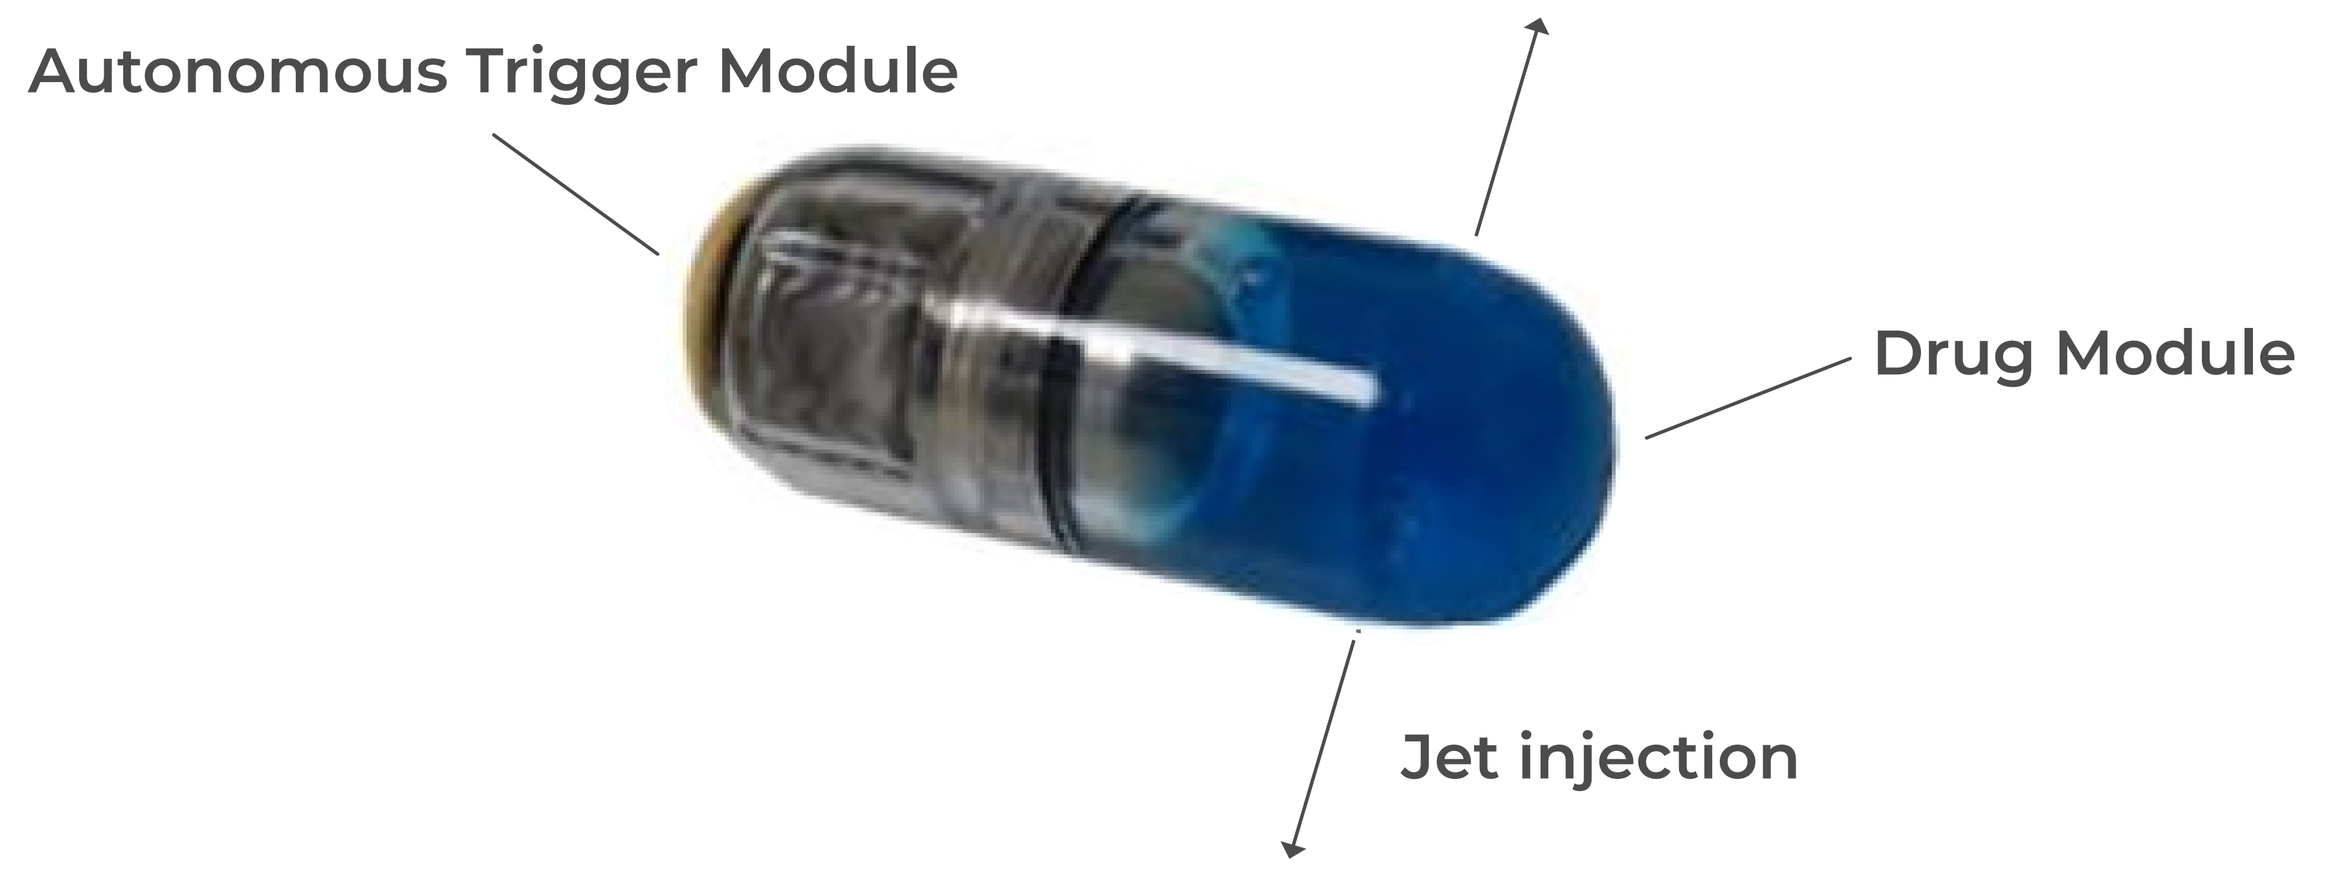
Autonomous Trigger Module
Drug Module
Jet injection

FIGURE 1. Image of autonomous OBDS capsule

OBJECTIVE

- Evaluate *ex-vivo* tissue ink deposition in both canine and swine tissue as compared to human tissue on bench.
- In parallel, evaluate the performance and assess the autonomous trigger function of the OBDS capsule *in vivo* in the canine model.

METHODS

Preclinical model development in *ex-vivo* small intestinal tissue

- Fresh swine tissue and fresh frozen tissue from human or canines was used for testing.
- The tissue was warmed to 37 °C, then placed on a 37 °C glass surface with the capsule placed inside of it. The trigger was submerged in 37 °C water. The tissue was kept moist with constant spraying of 37 °C water.
- After deployment, the tissue was cut open and excess ink rinsed out of the lumen. Ink deposition on *ex-vivo* small intestinal tissues of canine vs. swine vs. human was then compared.

In-vivo preclinical model development in canine

- The canine model was used to understand the *in-vivo* autonomous trigger functionality of OBDS due to similar GI transit and motility to humans **(see poster #106 for preclinical model selection)**.
- Two versions of an autonomous OBDS device (Type 1 and 2) were loaded with the radiopaque contrast reagent iohexol (OMNIPAQUE™ 350), and orally dosed to fasted male beagle dogs (N=12) with pentagastrin pre-treatment for fluoroscopic imaging.
- Two or more orthogonal images of each animal were collected prior to dosing, immediately after dosing, and 15-30 minutes post-dose to visualize capsule deployment time and location in the intestine.

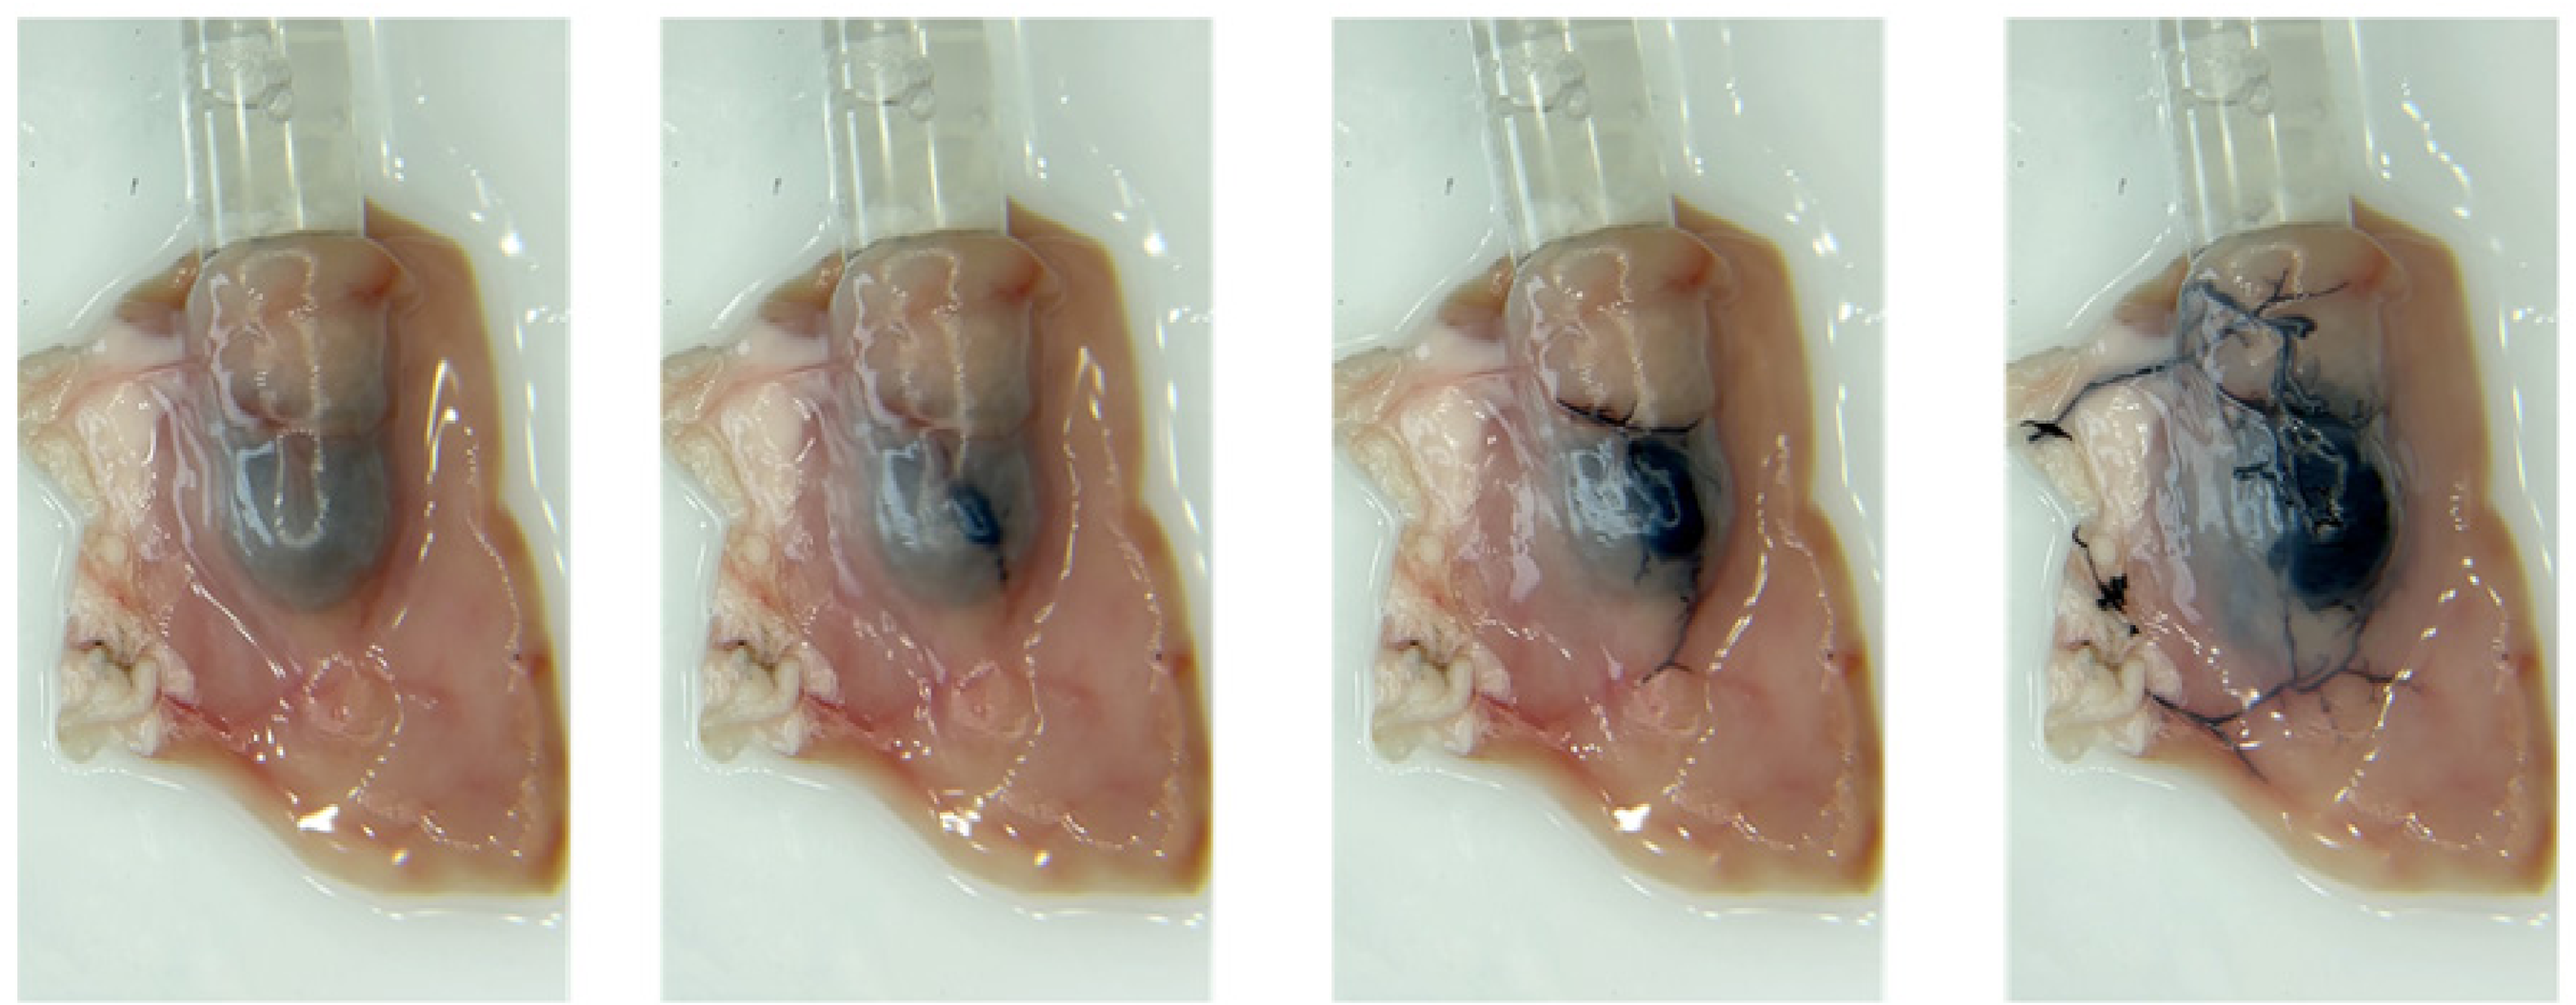

FIGURE 2. Time-lapse images of the *ex-vivo* trigger and deposition of OBDS ink device in swine small intestine tissue

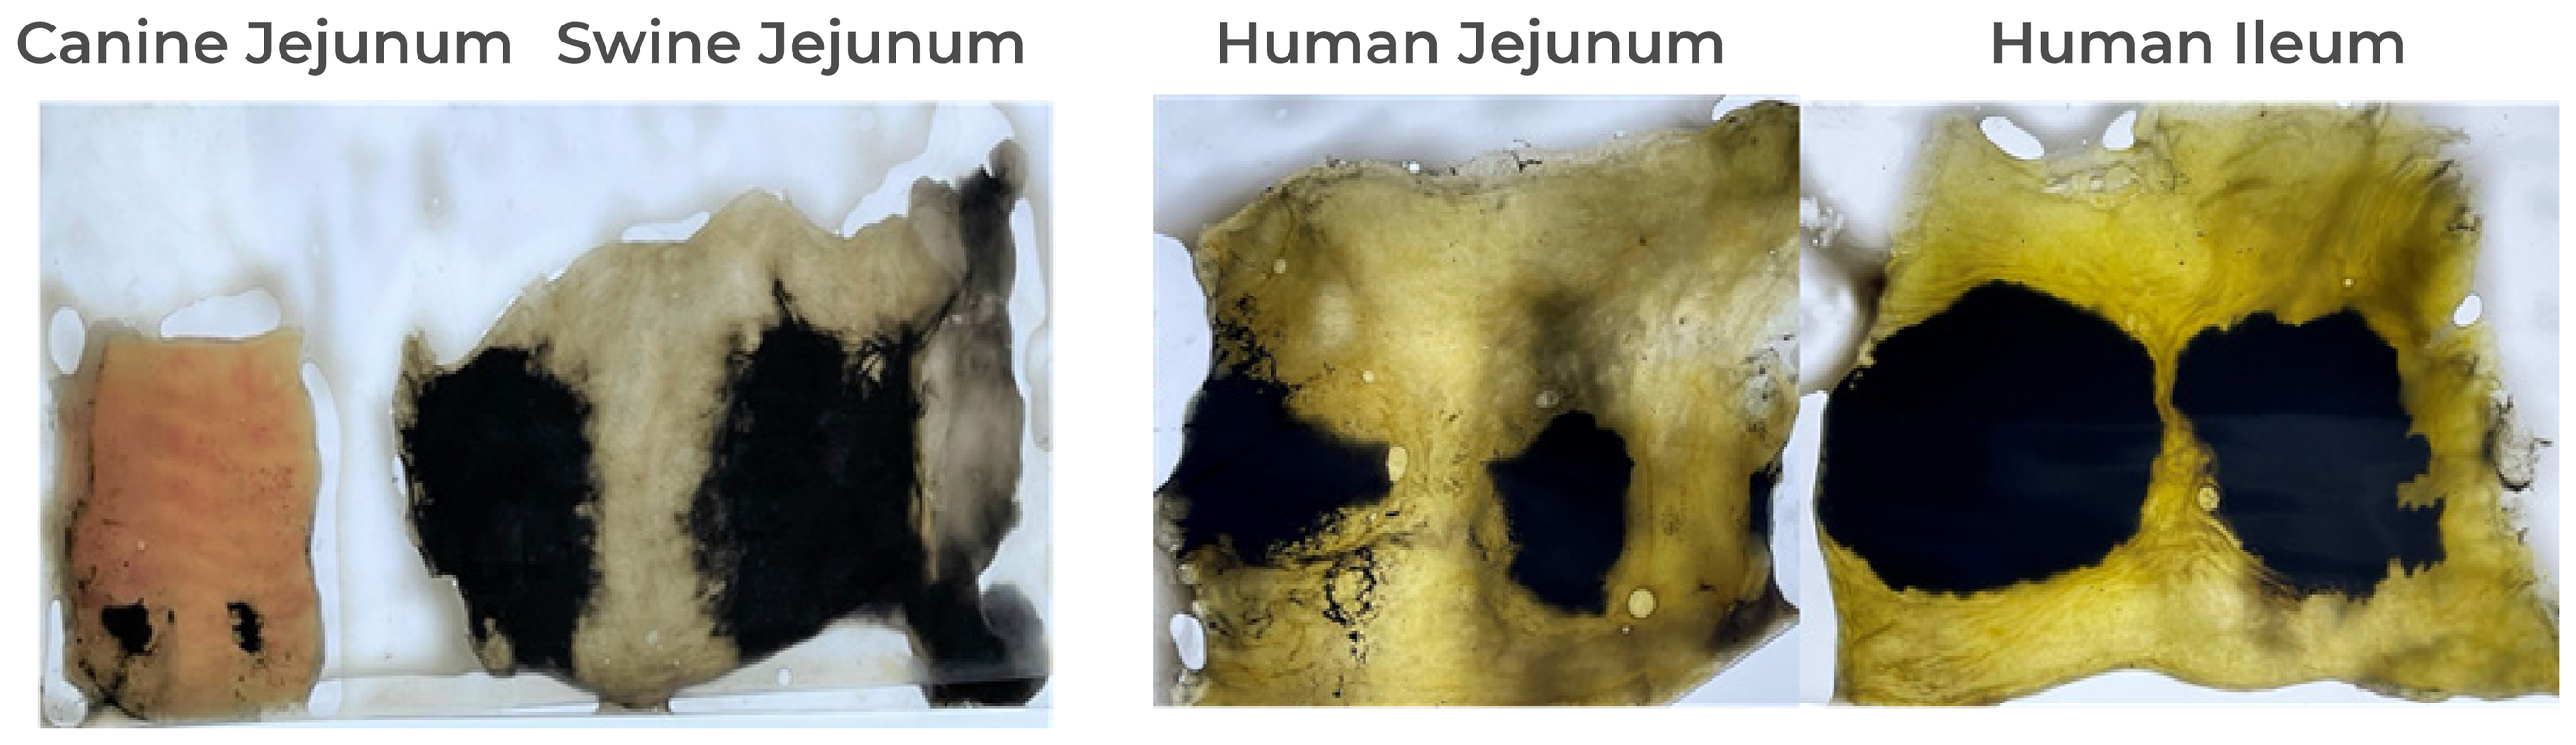
Canine Jejunum
Swine Jejunum
Human Jejunum
Human Ileum

FIGURE 3. *Ex-vivo* tissue deposition of India ink in intact bowel tissue in canine vs swine vs human tissue

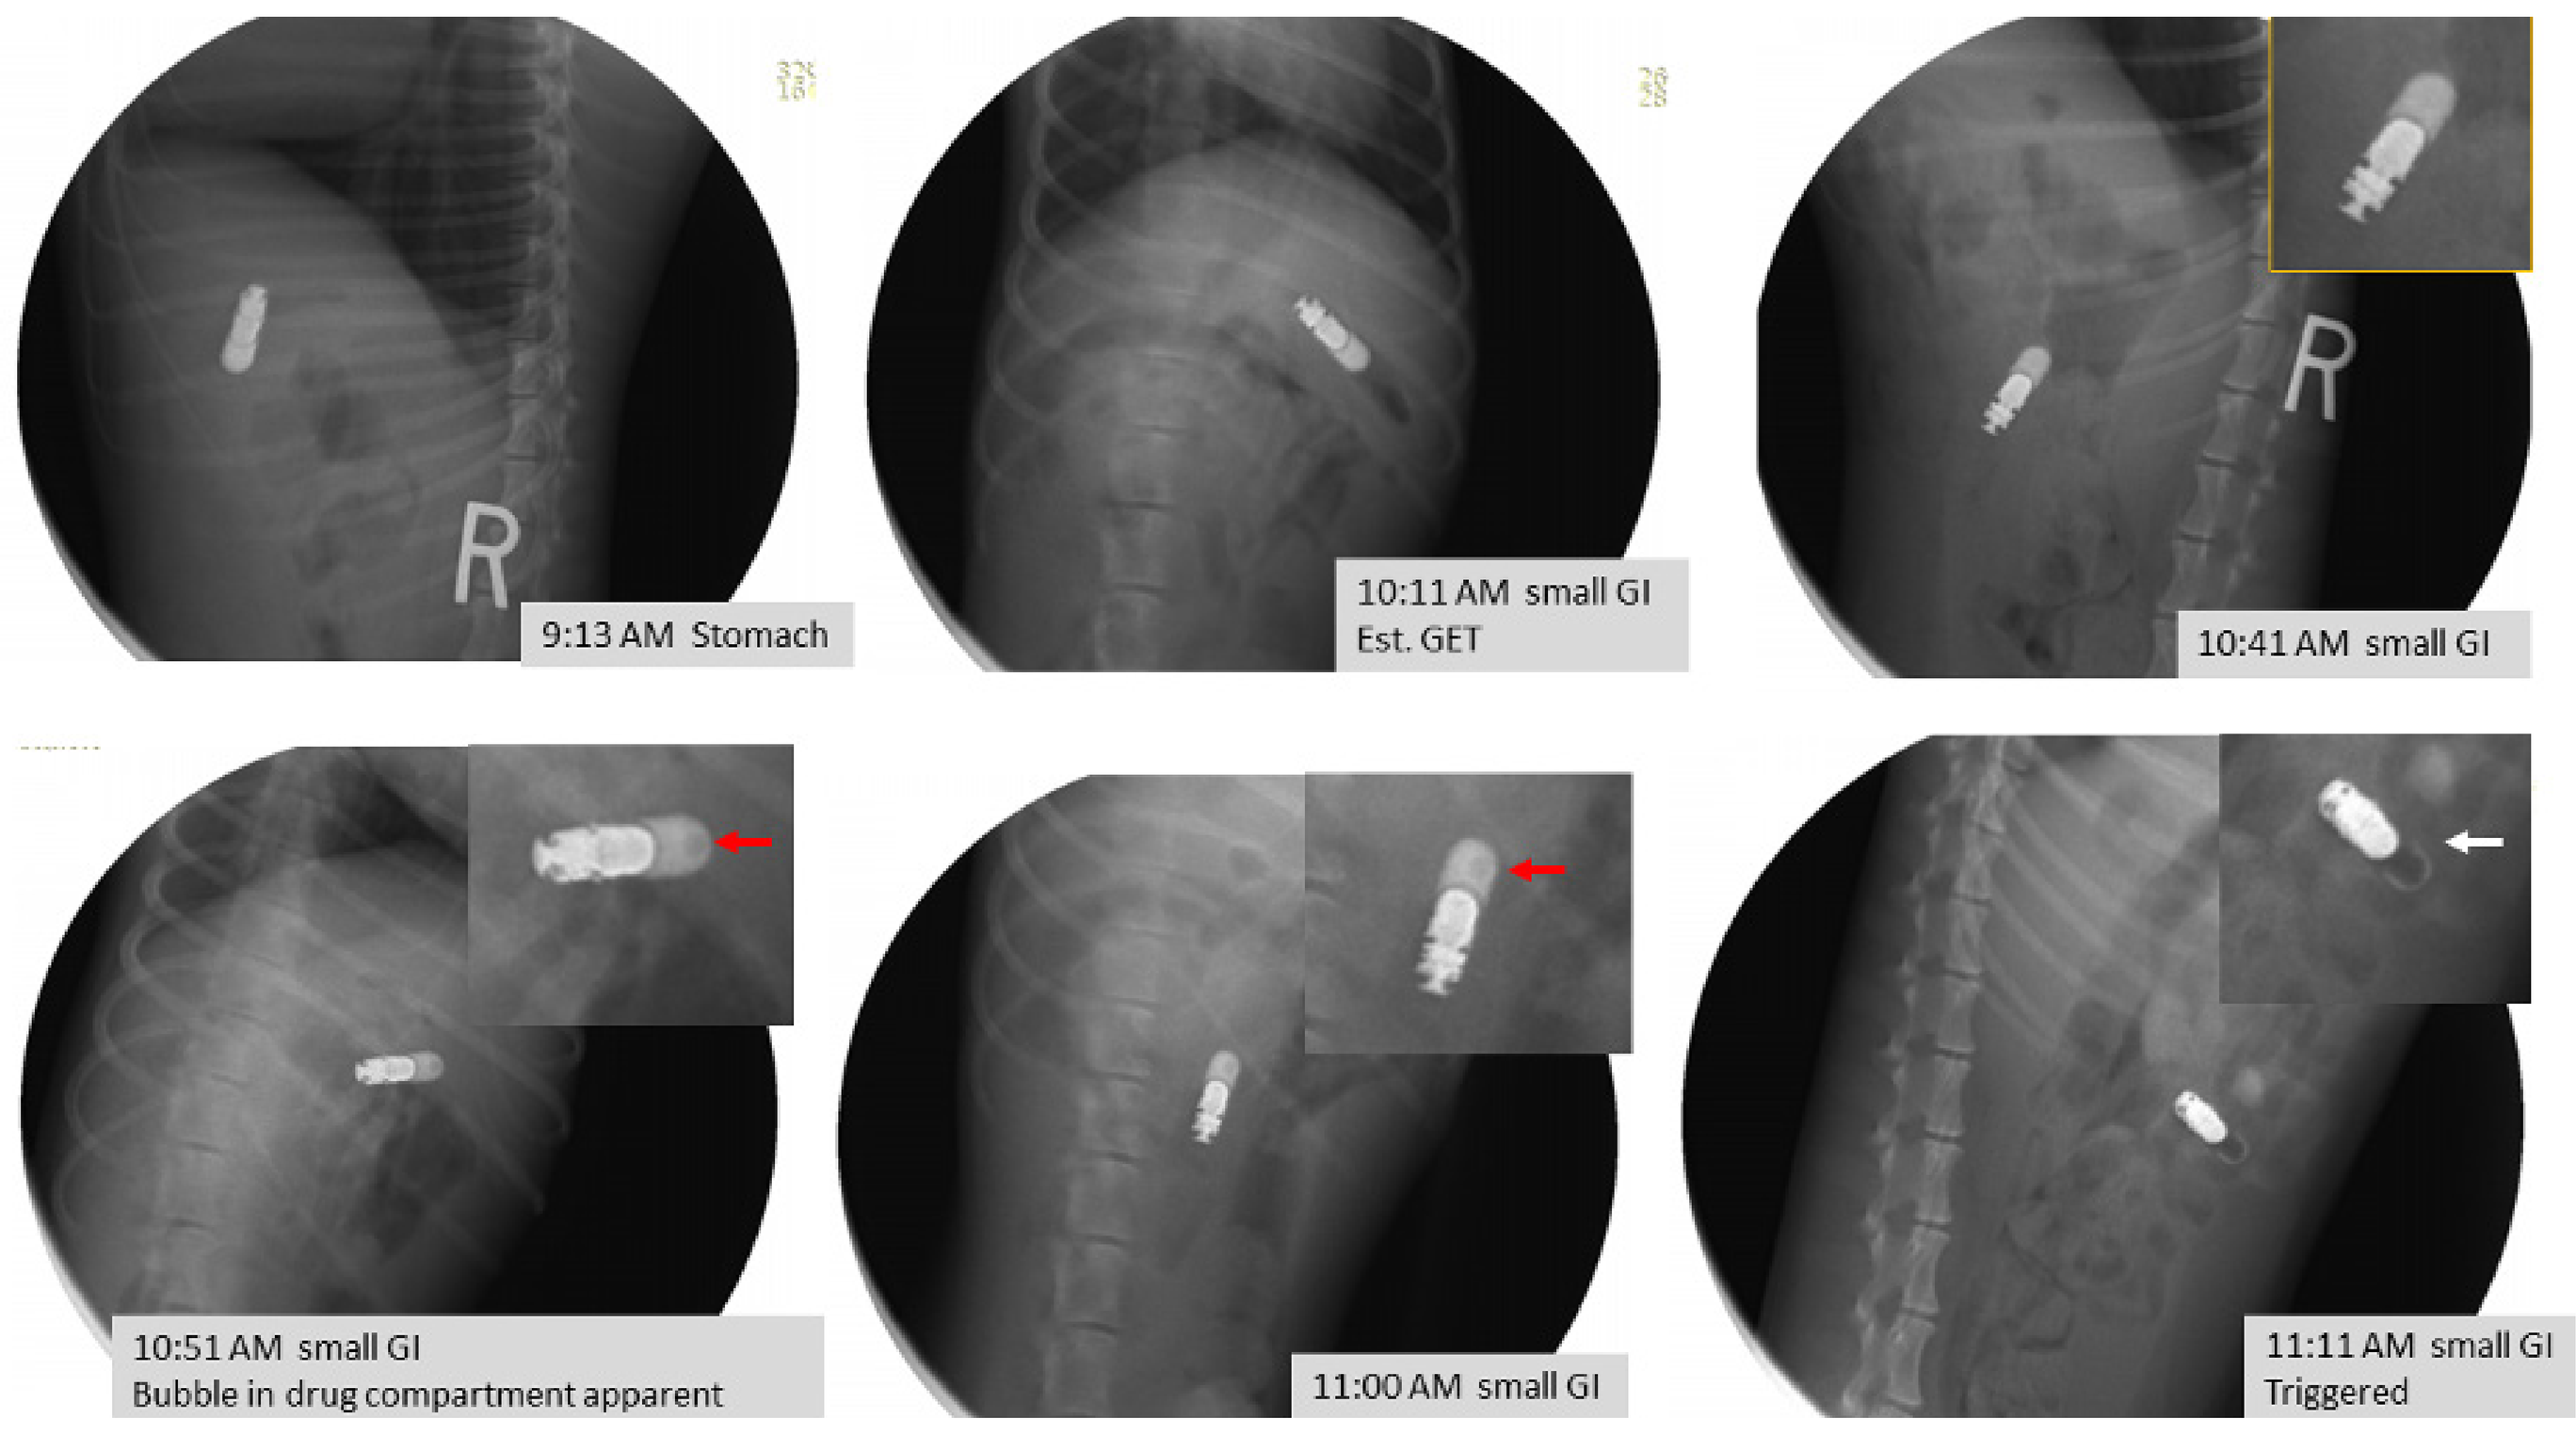
9:13 AM Stomach
10:11 AM small GI
Est. GET
10:41 AM small GI
10:51 AM small GI
Bubble in drug compartment apparent
11:00 AM small GI
11:11 AM small GI
Triggered

FIGURE 4. Representative image of capsule deployment in the small intestine. The drug module is noted with the red arrow. The layer of tungsten on the piston is noted by the white arrow.

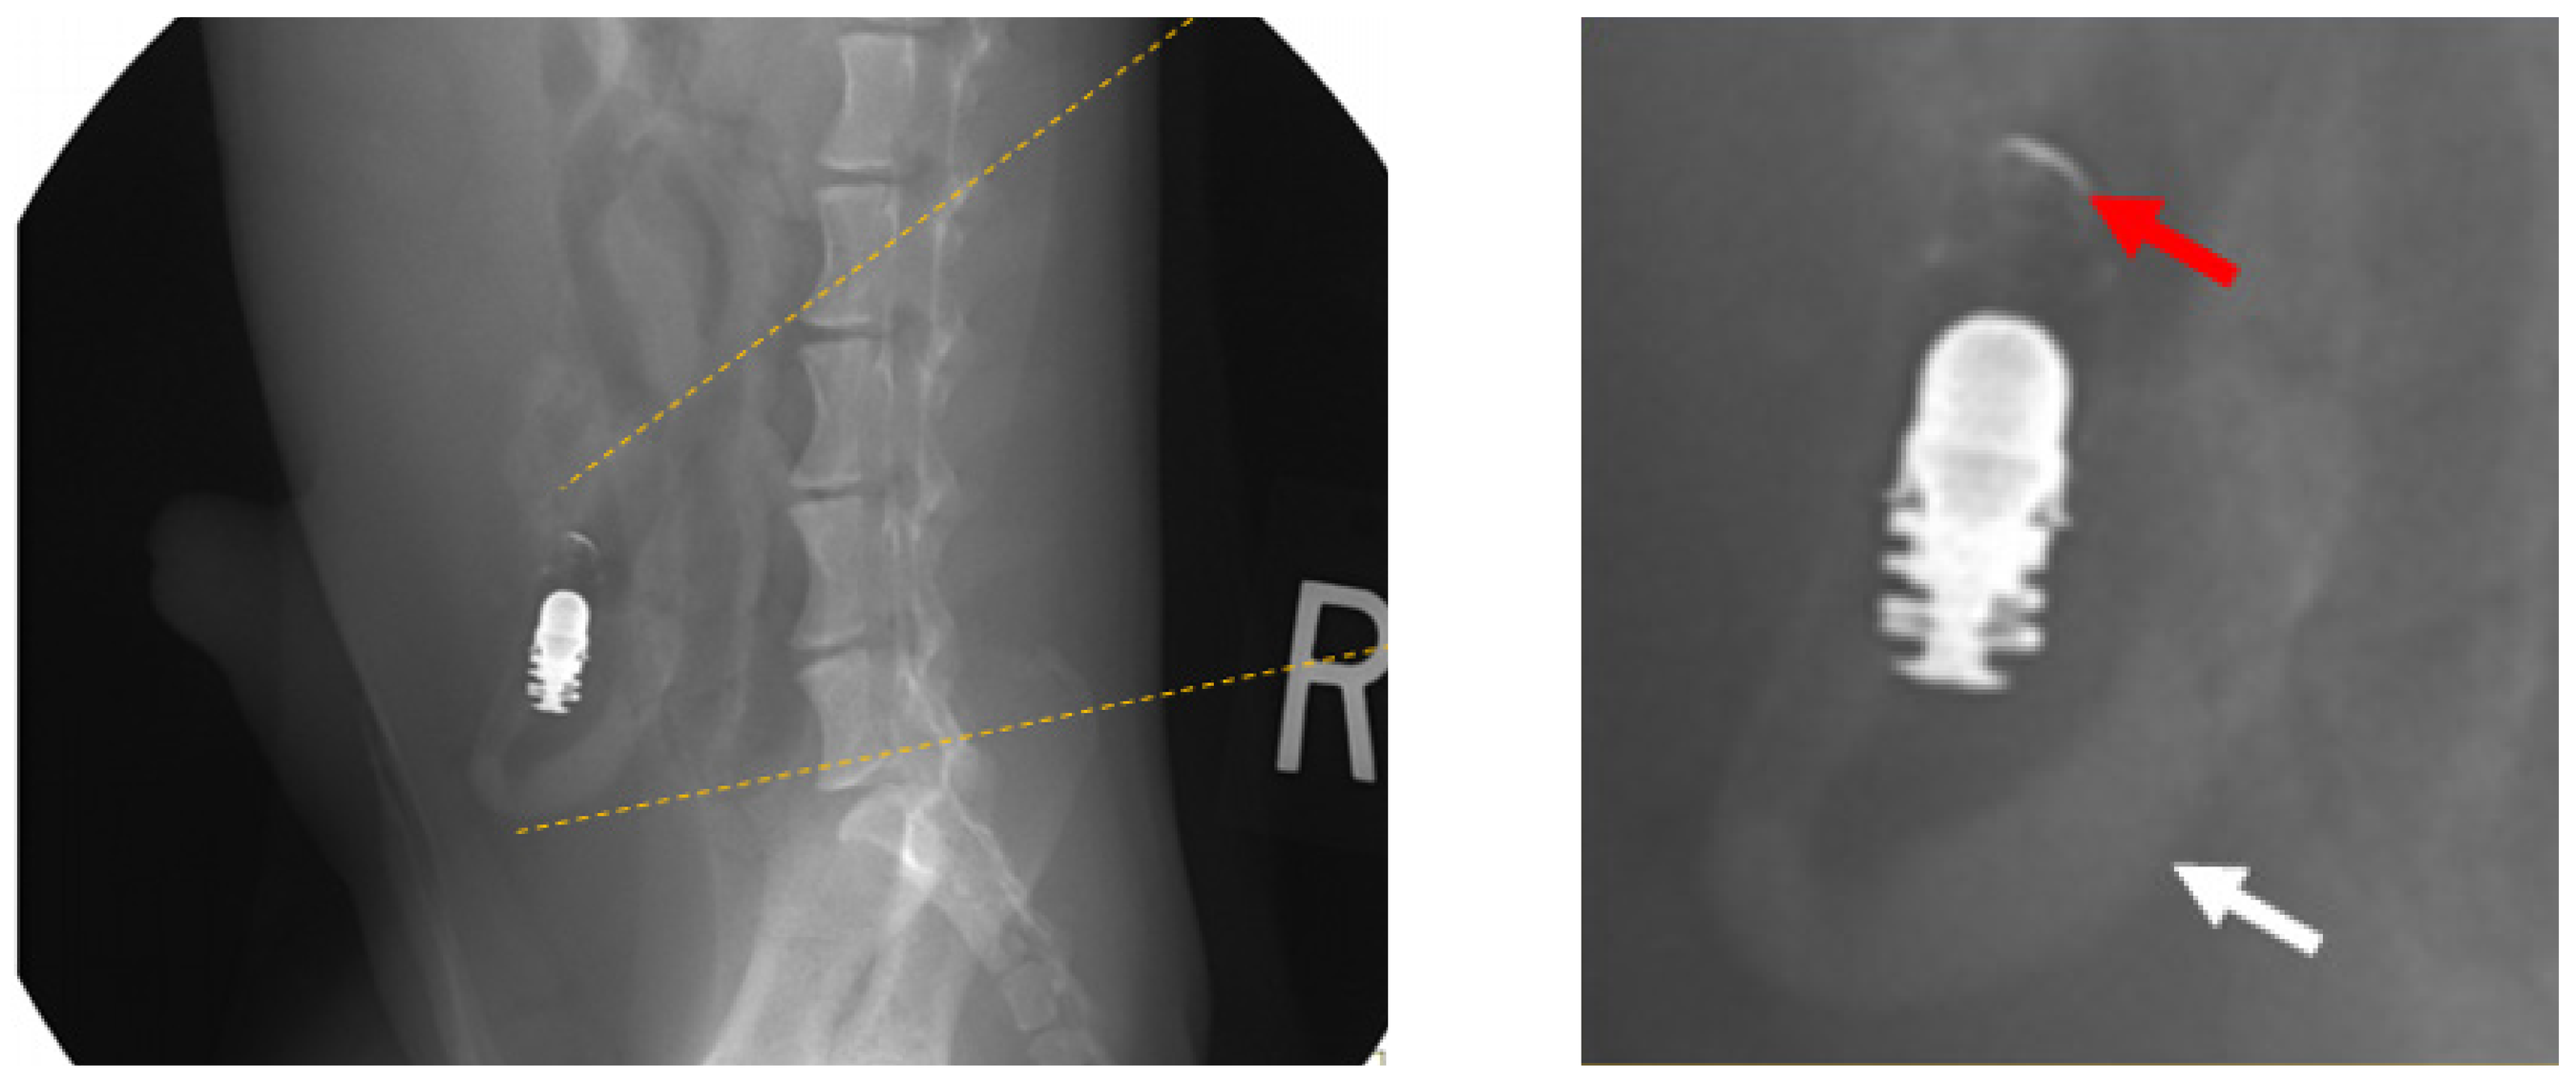

FIGURE 5. Representative image of small intestine post iohexol injection. Fully advanced piston is observed by placement of tungsten noted with the red arrow. Contrast staining of the intestinal wall post needless injection is noted by the white arrow.

RESULTS

Ex-vivo tissue ink deposition

- Significant ink deposition and distribution on the tissue were observed without penetrating the gut wall **(Figure 2)**.
- Ink deposition on the proximal small intestine tissue of swine and humans were similar.
- Lower and less consistent ink deposition on the canine proximal small intestine tissue was observed compared with humans **(Figure 3)**.
- This suggests a lower capacity of canine tissue to accept a liquid bolus/deposition as expected due to differences in canine intestinal anatomy compared to humans and swine.

In-vivo trigger performance

- We observed 10 out of 12 capsules dosed orally deployed approximately at the small intestine. One deployed in the colon, and one did not deploy due to manufacturing defects **(Figure 4)**.
- Deployment was indicated by the observation of advancement of the piston and the disappearance of iohexol from the capsule **(Figure 5)**.
- The overall triggering time was consistent in each group.
- Type 2 capsules had an average deployment of 1 h and 8 min ± 5 min post gastric emptying (N=7) which was ~14 minutes faster than Type 1 capsules (N=4) **(Figure 6)**.
- Three animals had rapid GI transit (5-7h), suggesting that earlier deployment of the device may be preferable to ensure deployment in the small intestine.

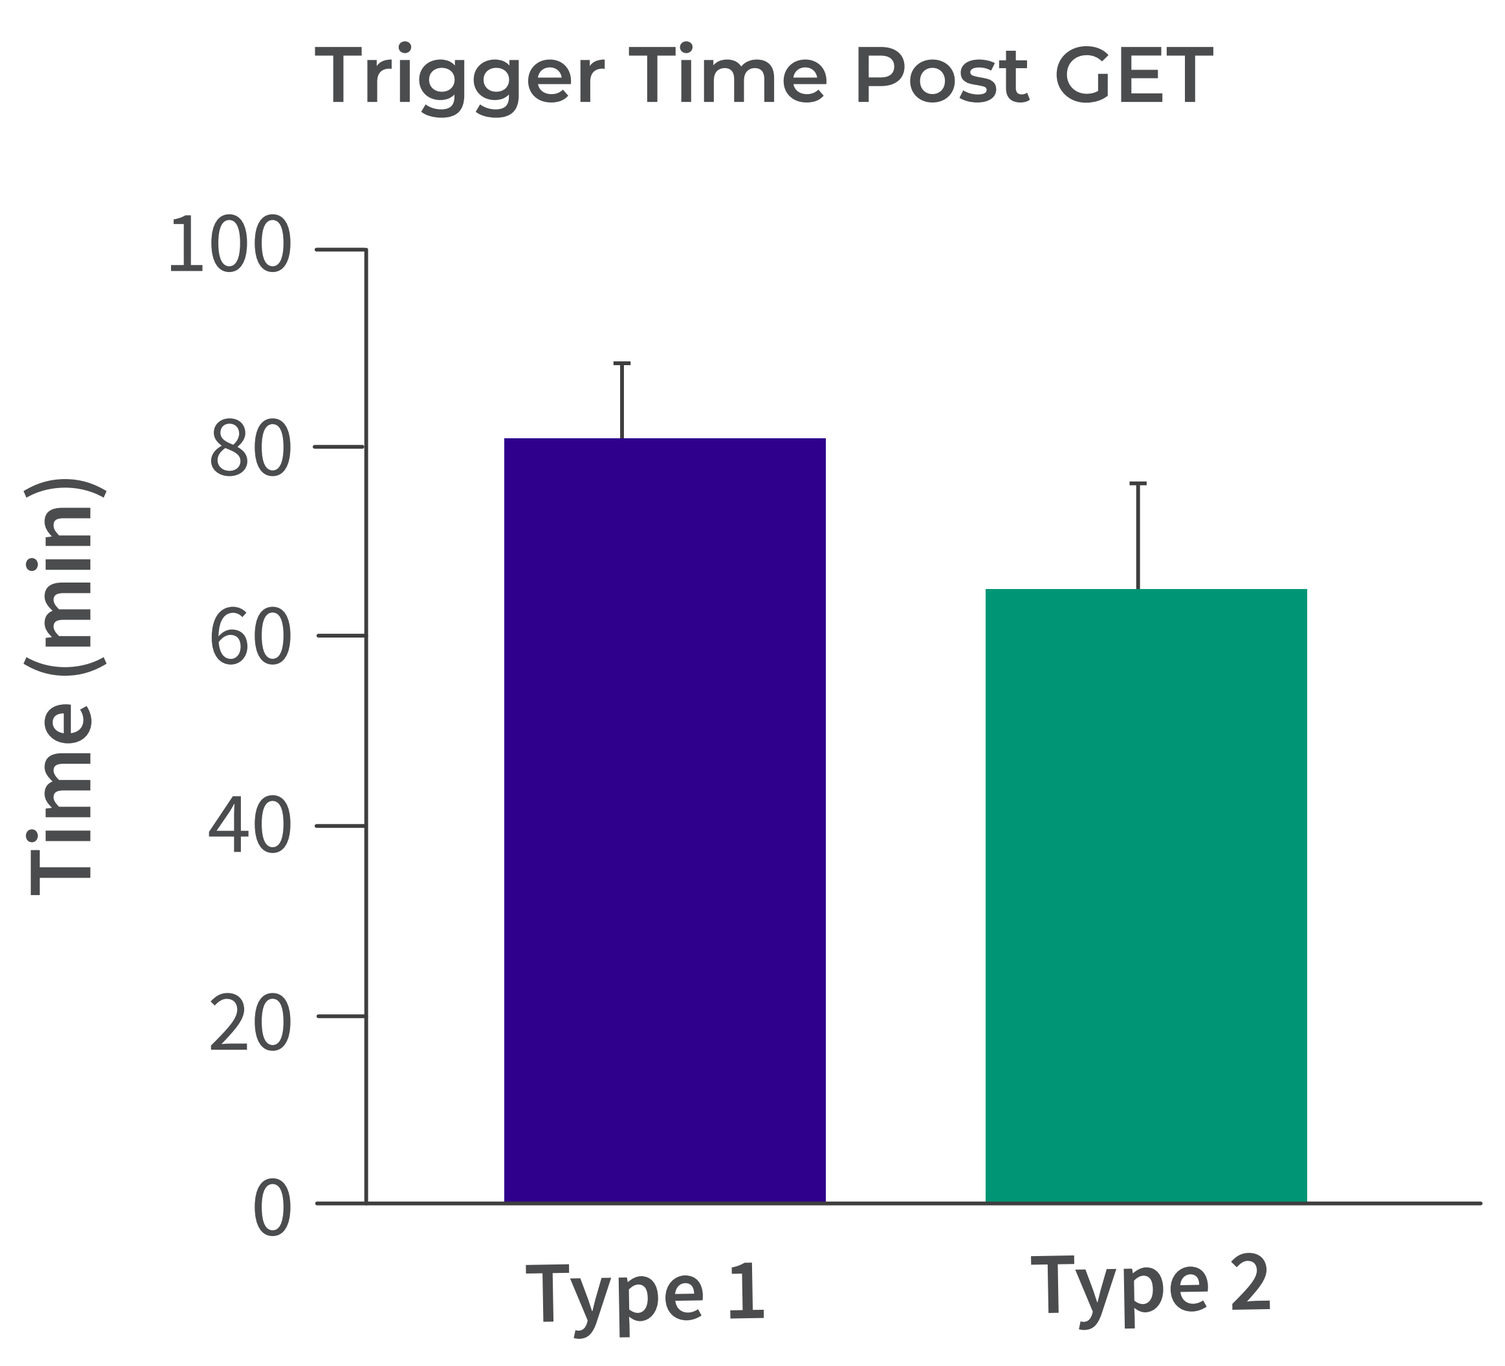
Trigger Time Post GET
Time (min)
0
20
40
60
80
100
Type 1
Type 2

Figure 6. Triggering time of Type 1 (N=4) and Type 2 (N=7) trigger post gastric emptying (GET).

SUMMARY AND DISCUSSION

- We have successfully demonstrated ≥ 83% deployment accuracy of autonomous OBDS capsules in the small intestine, and consistent deployment time post gastric emptying without early deployment in the stomach.
- These results suggest the canine model can be utilized to assess consistency and reproducibility of the OBDS capsule, and triggers can be modified to adjust small intestinal deployment time.
- Assessment of submucosal injection and absorption should be performed in the swine model for human translation of the oral bioavailability of OBDS capsules, due to less injection volume/deposition in canine tissue (see poster#106).

Scan the code to view an electronic version of this poster, or visit biioratherapeutics.com/publications

© 2022 Biora Therapeutics, Inc. All rights reserved. Biora Therapeutics™ is a trademark of Biora Therapeutics, Inc.

bioratherapeutics.com

Poster #105
Presented at Controlled Release Society Annual Meeting, July 13-14, 2022

CONTACT INFO
Chris Wahl, MD, MBA
VP, Strategy and Business Development
chris.wahl@bioratherapeutics.com