Development of a submucosal injection device for an oral biotherapeutic delivery system

Shaoying Nikki Lee[1], Cheryl Stork[1], Jeff Smith[1], Bryan Smith[1], Nelson Quintana[1], Chris Wahl[1], Sharat Singh[1]

1. Biora Therapeutics Inc., CA, USA

INTRODUCTION

Biologics/peptides/nucleic acids are highly effective drugs, however, oral delivery of these therapeutics has proven difficult due to the harsh conditions of the upper gastrointestinal tract (GIT) and the poor absorption rate in the small intestinal mucosa. The current state of-the-art technology for a successful oral protein delivery provides around 1% bioavailability when delivered as an oral tablet (Rybelsus® oral Semaglutide).

We aim to develop an oral biotherapeutic delivery system (OBDS) that prevents drug degradation in the upper GIT and increases oral bioavailability via liquid jet delivery to the small intestine using an ingestible device.

THE OBDS DEVICE

- The autonomous OBDS device is comprised of a drug module, which houses a formulation of a therapeutic compound, and a trigger module under pressurized gas control **(Figure 1)**.
- The OBDS device operates autonomously and provides a needleless liquid jet injection to deposit the liquid drug payload into the submucosal space of the proximal small intestine for absorption into systemic circulation.

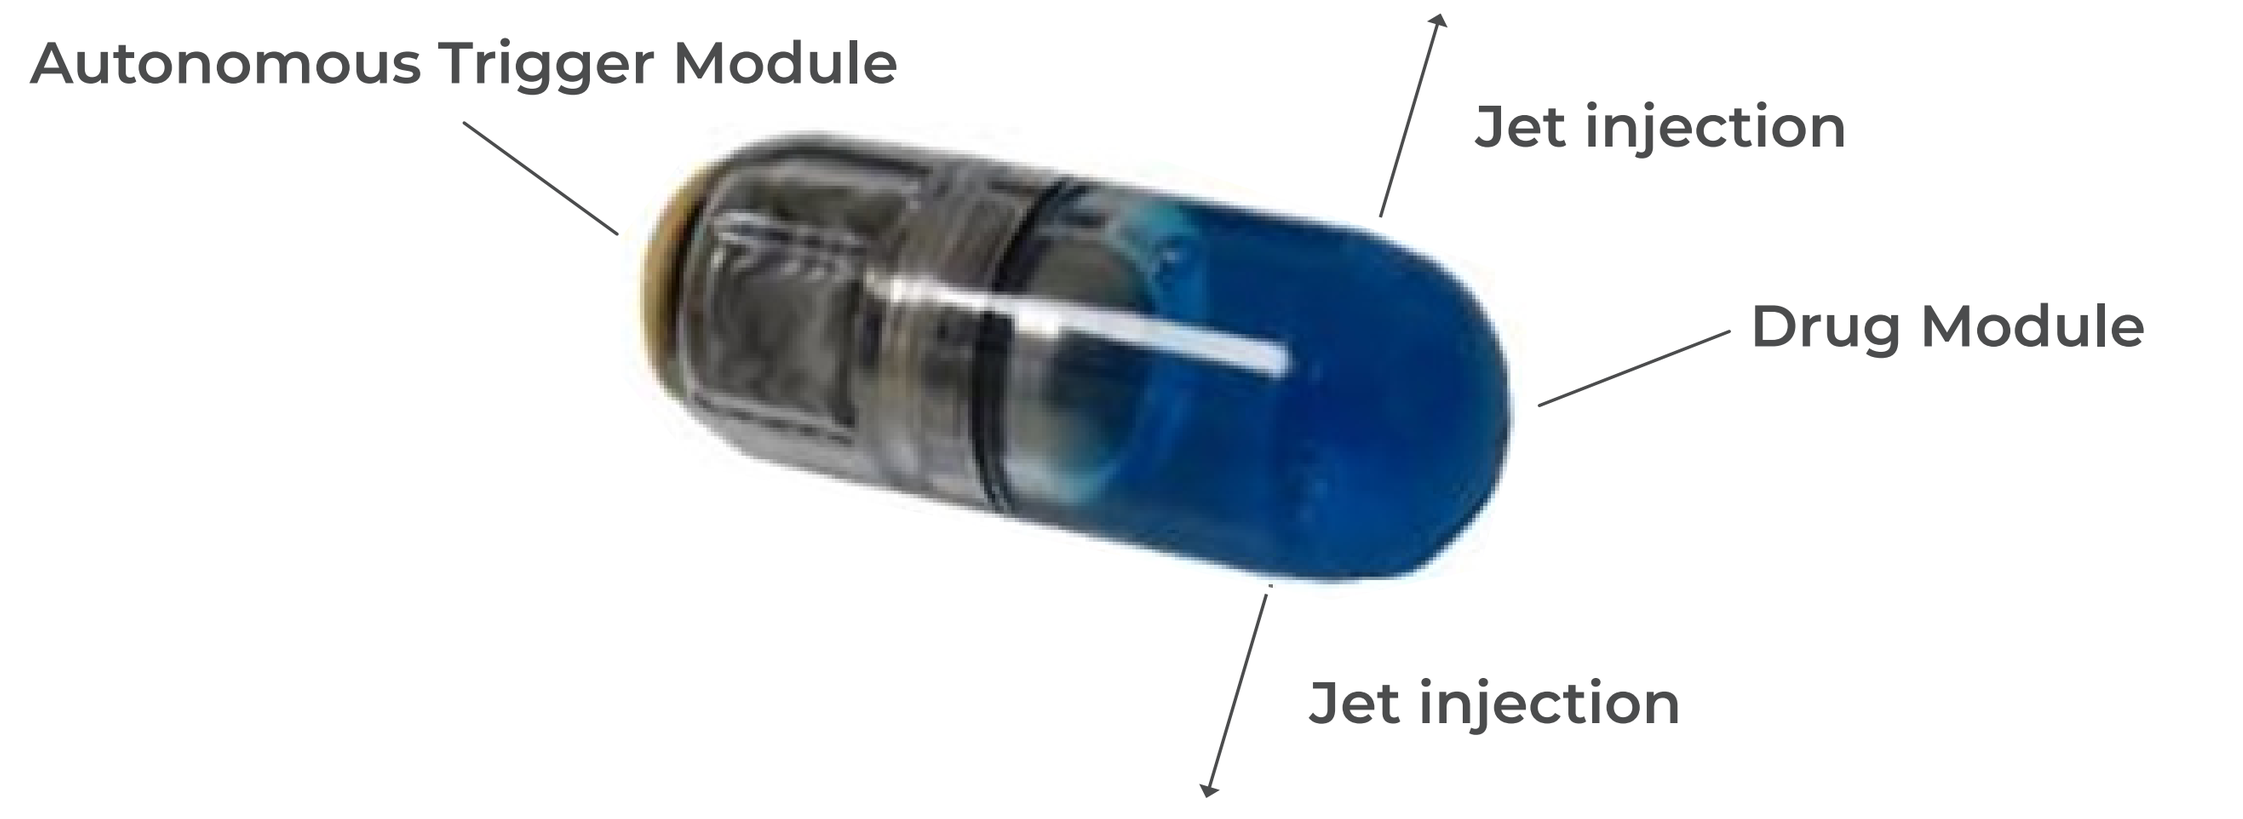
Autonomous Trigger Module
Jet injection
Drug Module
Jet injection

FIGURE 1. Image of autonomous OBDS device

OBJECTIVE

To evaluate the *in vivo* performance of the OBDS device using:

- An oral dose beagle dog model to evaluate autonomous trigger function.
- An intraduodenal endoscopic delivery swine model to demonstrate potential submucosal injection efficiency for human translation.

Scan the code to view an electronic version of this poster, or visit biioratherapeutics.com/publications

METHODS

Preclinical animal model selection for human translation (Figure 2)

- The canine model was selected for the evaluation of repeatability and safety of the autonomous device due to its similar GI transit and motility to human **(Table 1)**.
- The swine model was selected for proof of MOA and performance due to similar anatomical and histological features to human **(Table 1)**.

TABLE 1: Physiological and anatomical differences between animal models and humans

	Gastric pH[a]	Duodenum pH[a]	Gastric Emptying Time, Fasted (hr)	Gastric Emptying Time, Fed (hr)	Small Intestinal Transit Time (hr)	Small Intestinal Volume Time (ml)[a]	Small Intestinal Villi Shape
Human	0.4 – 4 (fasted) 2 – 4.5 (fed)	5 – 7	0.66 – 1[b]	2 – 5[g]	2 – 4[a]	212 ± 110	Finger[a]
Swine	1.4 – 4 (fasted) 4.4 (fed)	6	Variable; 1.4 and up to 20 days[c,d]	Variable[h,j]	Variable 3 – 4; 1 – 2 days[d,h]	476 ± 253	Finger[a]
Canine	1.5 (fasted) 3 – 5 (fed)	6.2	0.4 – 1[e,f]	Variable; 12 – 13[j]	2 – 3[j]	300	Long and slender[a]

[a]Hatton et al. 2015; [b]Worsoe et al. 2011; [c]Davis et al. 2001; [d]Hossain et al 1990; [e]Mahar et al 2012; [f]Kolziek et al 2019; [g]Lee et al 2014; [h]Gregory 1990; [i]Treacy 1990; [j]Linbury et al 2012

In-vivo preclinical model development in canine

- Two versions of an autonomous OBDS device (Type 1 and 2) were loaded with the radiopaque contrast reagent iohexol (OMNIPAQUE™ 350), and orally dosed to fasted male beagle dogs (N=12) with pentagastrin pre-treatment for fluoroscopic imaging.
- Orthogonal images of each animal were collected prior to dosing, immediately after dosing, and 15-30 minutes post-dose to visualize device deployment time and location in the intestine **(Figure 3)**.
- Deployment was indicated by the observation of advancement of the piston and the disappearance of iohexol from the device **(Figure 3)**.

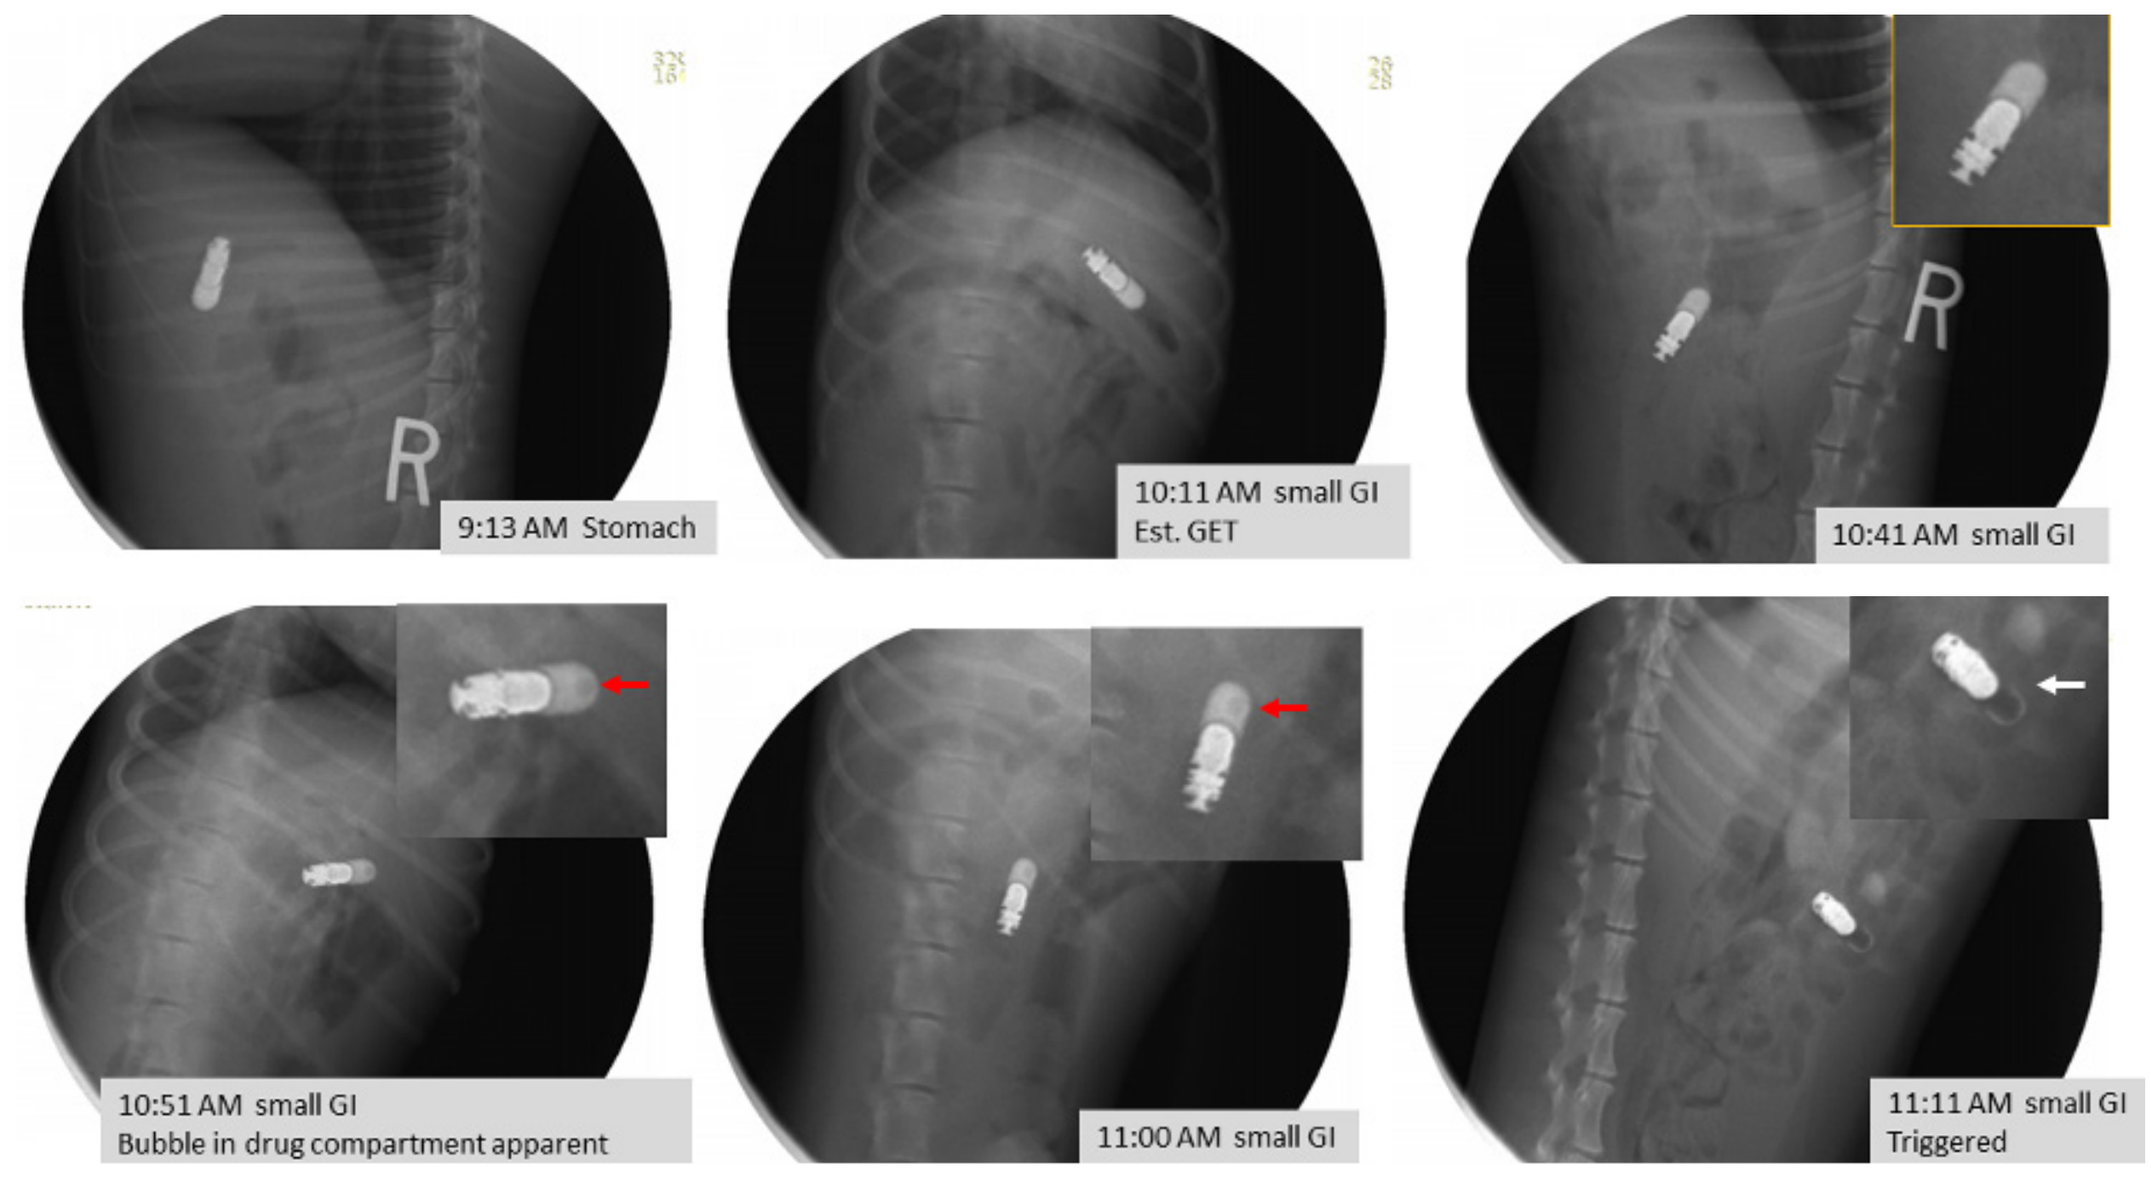
9:13 AM Stomach
10:11 AM small GI
Est. GET
10:41 AM small GI
10:51 AM small GI
Bubble in drug compartment apparent
11:00 AM small GI
11:11 AM small GI
Triggered

FIGURE 3. Representative images of device deployment in the small intestine.
The drug module is noted with the red arrow. The layer of tungsten on the piston is noted by the white arrow.

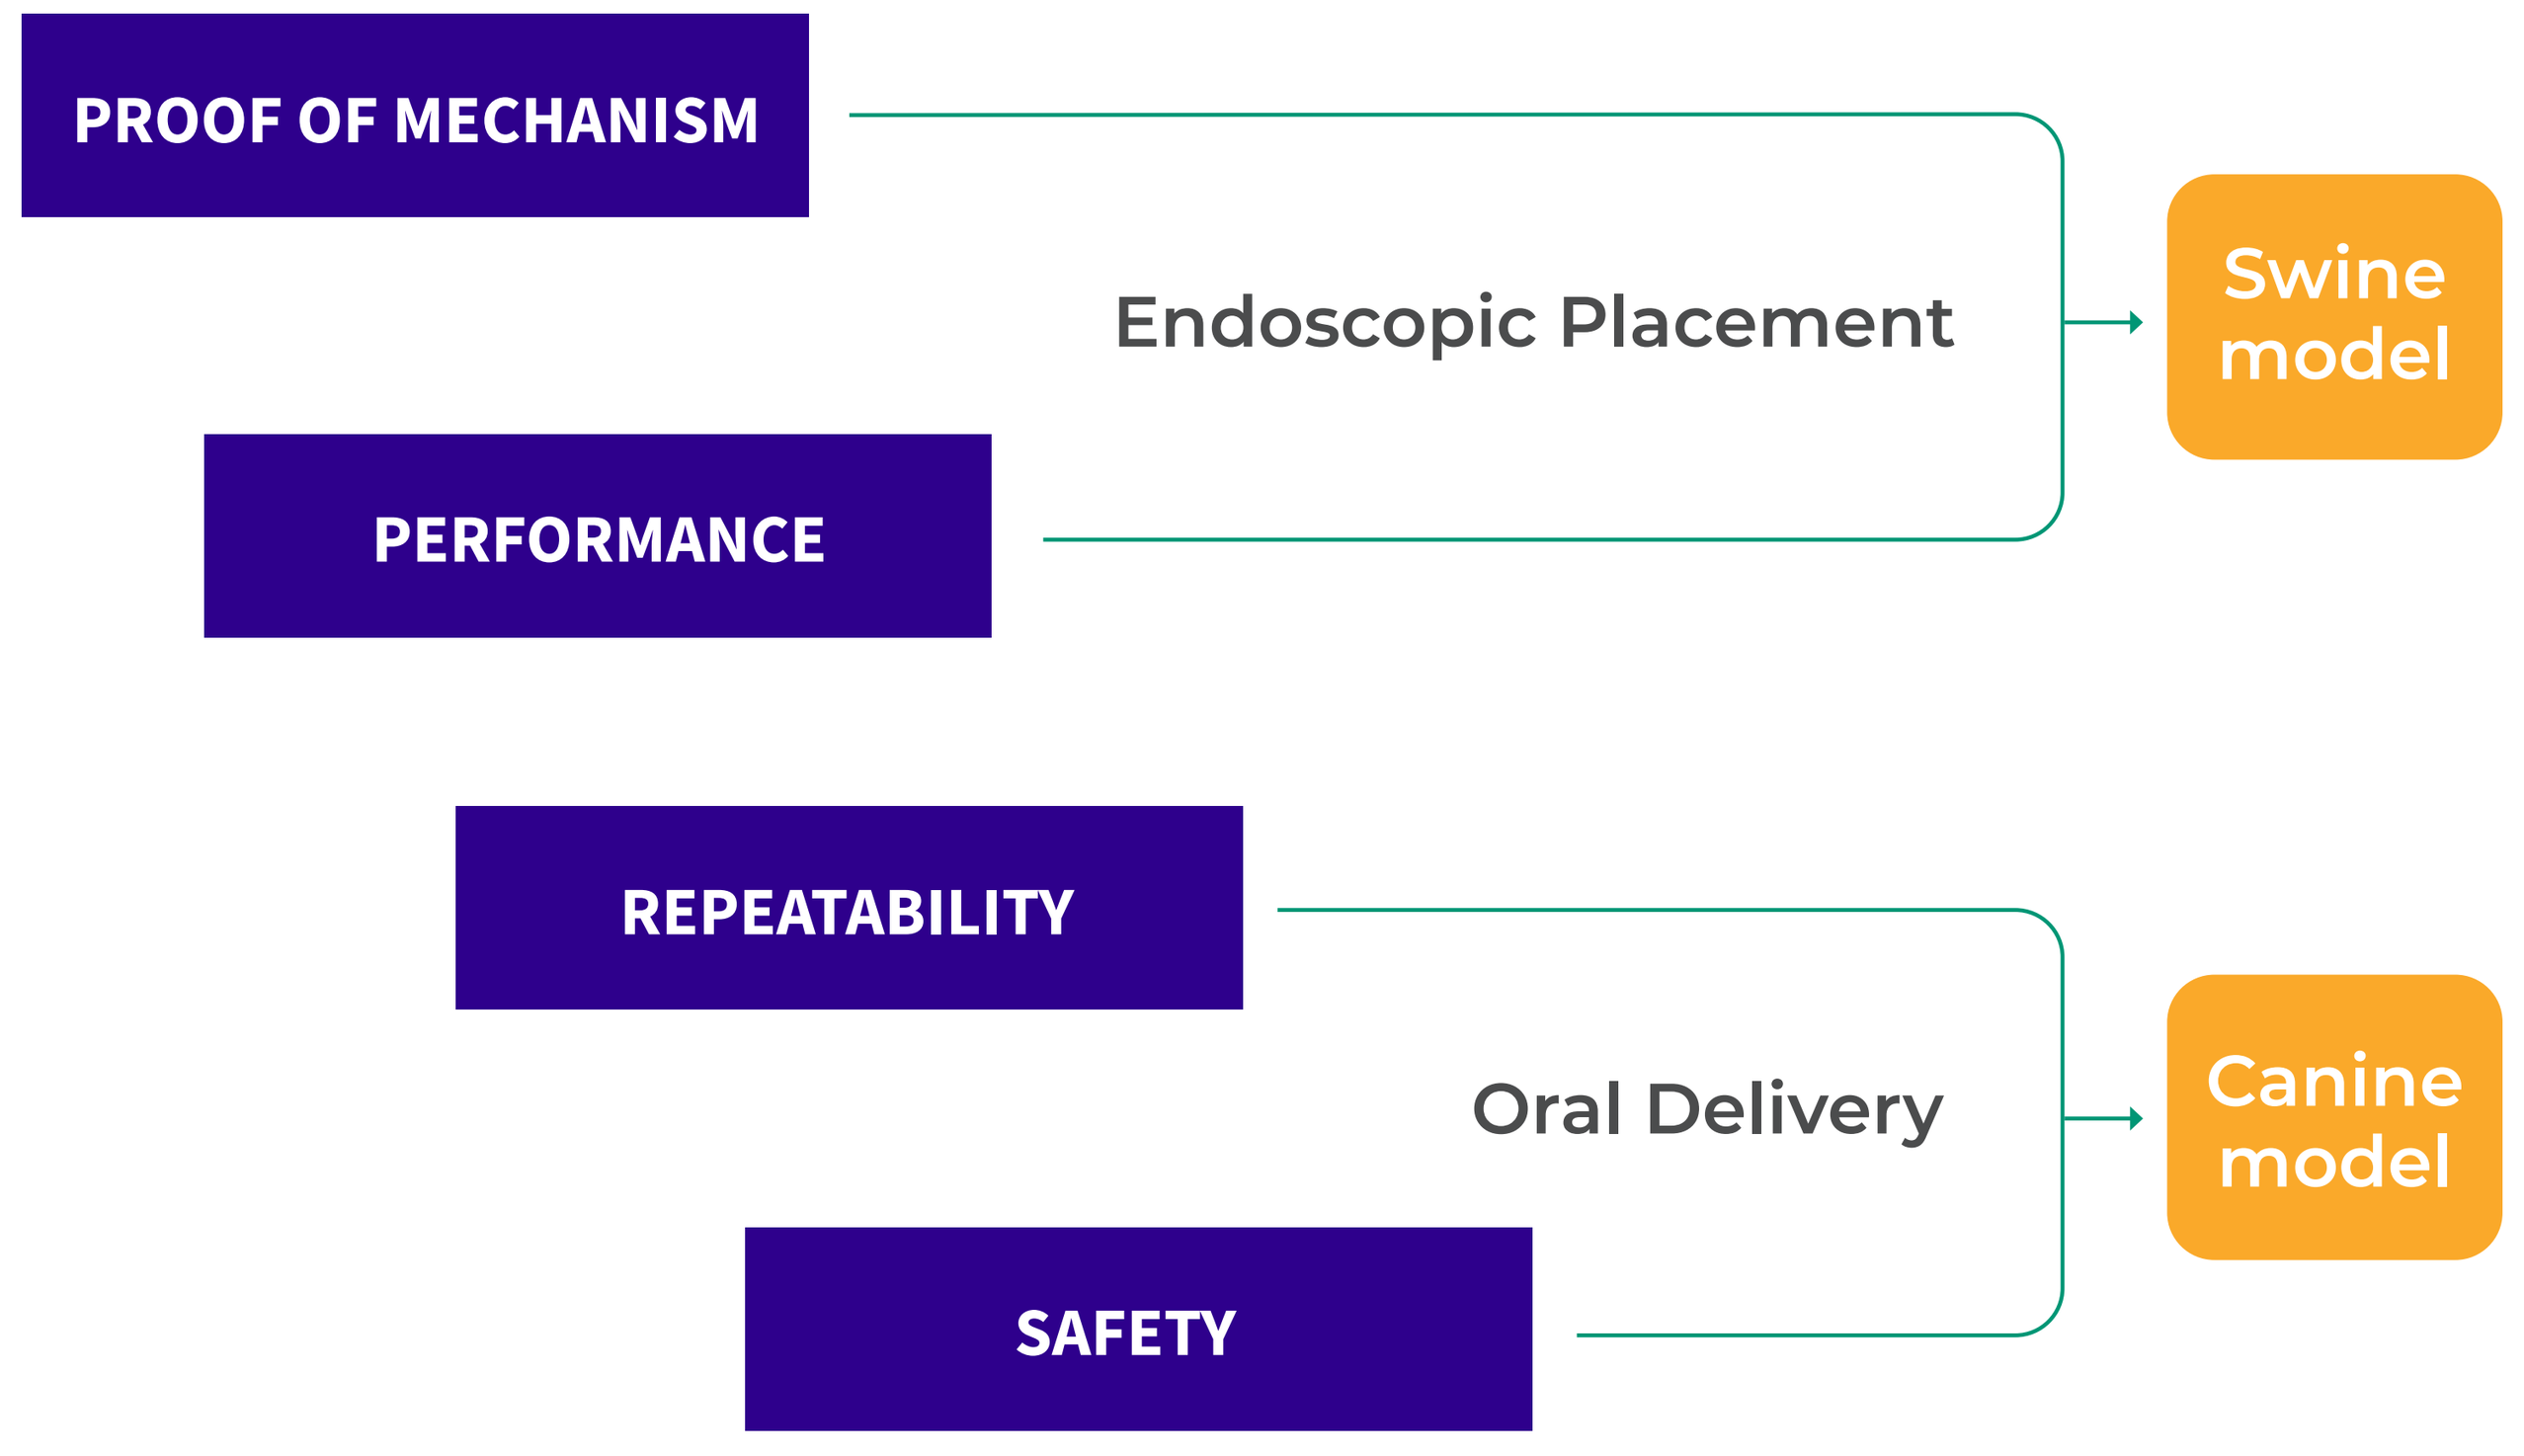
PROOF OF MECHANISM
PERFORMANCE
Endoscopic Placement
Swine model
REPEATABILITY
SAFETY
Oral Delivery
Canine model

FIGURE 2. Preclinical Model Selection

Intraduodenal (ID) endoscopy placement of the OBDS in swine

- OBDS devices filled with a variant of adalimumab were attached to the endoscope via the working channel and endoscope delivery device and inserted orally into fasted animals under anesthesia.
- The device was advanced past the pyloric sphincter and expelled from the placement instrument in the proximal small intestine. The device then naturally transited and autonomously triggered in the GIT.
- Blood samples post-ID dosing were collected to evaluate the injection efficiency of the OBDS compared with the IV control group.

In-vivo OBDS-ink deposition in swine

- OBDS devices filled with India ink were placed in the proximal small intestine and manually triggered **(Figure 4, A-C)**.
- At 24 hours post-dose, the animal was sacrificed, and ink deposition was observed in the small intestine **(Figure 4, D)**.

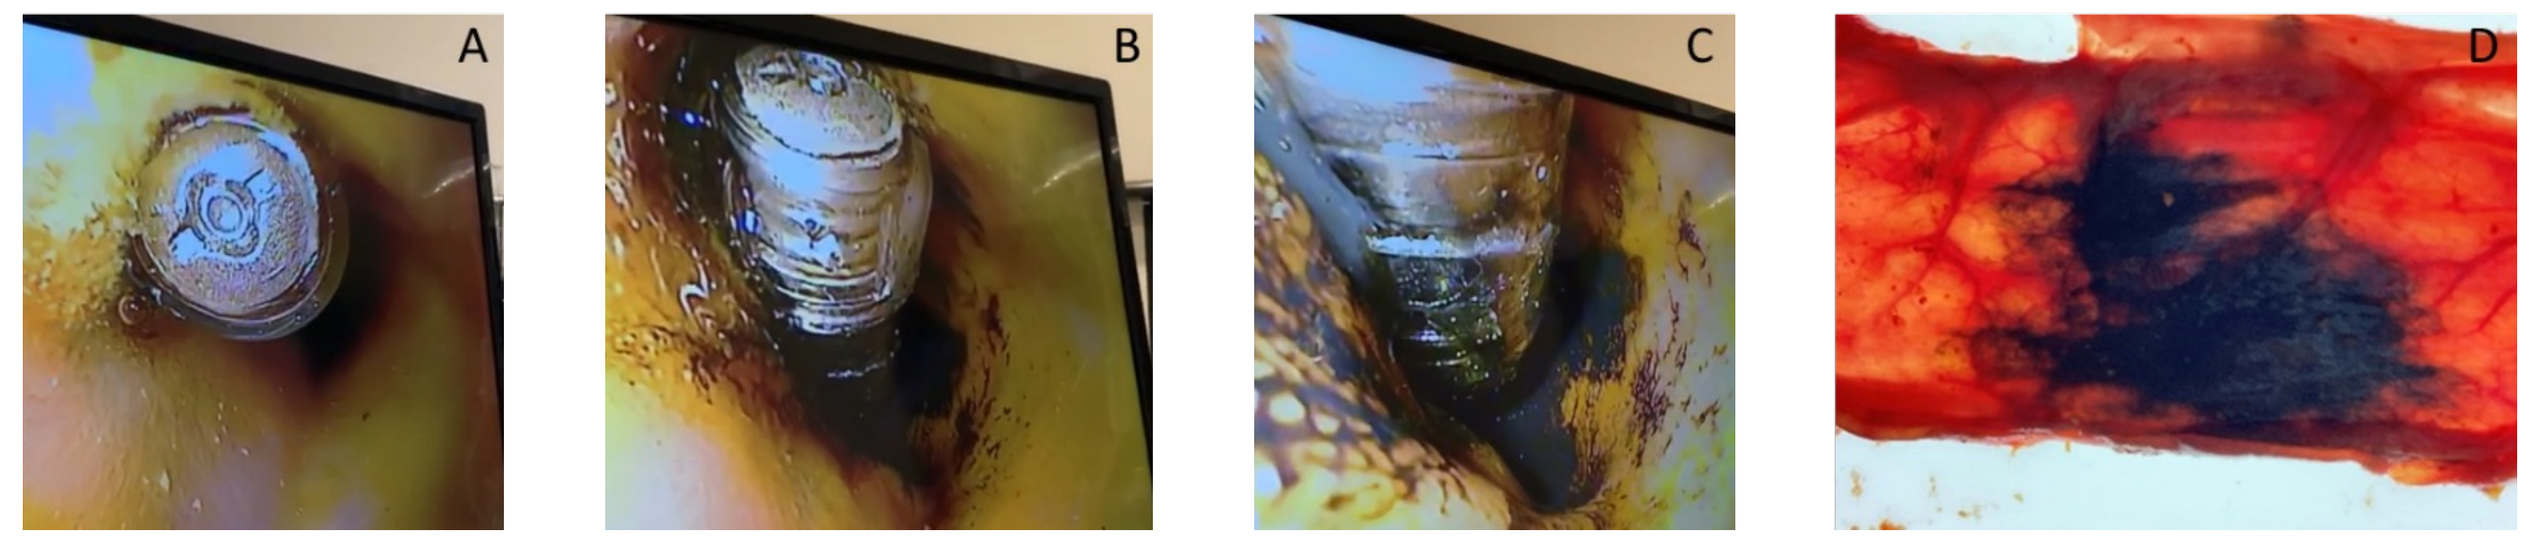
A
B
C
D

FIGURE 4. Endoscopy placement of OBDS ink device *in vivo*.
A. Placement of OBDS device in the duodenum; B. manual triggering of device; C. Close-up look at the tissue ink deposition; D. Ink deposition at terminal necropsy of swine duodenum at 24hr post-deployment.

RESULTS

In vivo trigger performance in beagle dogs

- 10/12 (~83%) of devices dosed orally deployed approximately at the small intestine. Type 2 devices had an average deployment of 1 h and 8 min ± 5 min post gastric emptying (N=7) which was ~14 minutes faster than Type 1 devices (N=4) **(Figure 5)**. Overall triggering time was consistent in each type of trigger.
- Three animals had rapid GI transit (5-7h), suggesting that earlier deployment of the device may be preferable to ensure deployment in the small intestine.

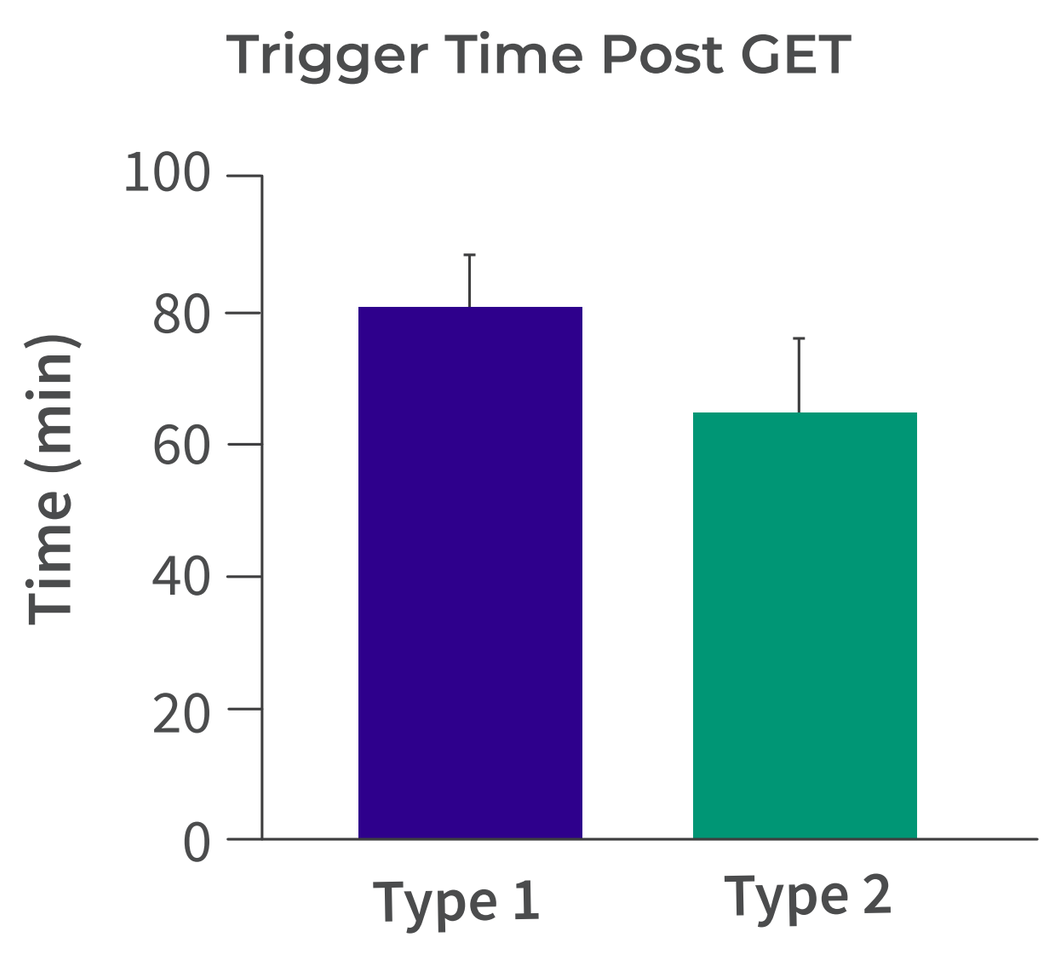
Trigger Time Post GET
Time (min)
100
80
60
40
20
0
Type 1
Type 2

Figure 5. Triggering time of Type 1 (N=4) and Type 2 (N=7) trigger post gastric emptying (GET).

Pharmacokinetics via endoscopic placement in swine

- All OBDS devices were successfully advanced through the pyloric sphincter, without early deployment, and were released in the proximal duodenum to naturally transit and deploy *in vivo.*
- Eight animals showed detectable drug levels **(Figure 6)** and an oral bioavailability average of 22% or 25% (range from 7-55%), excluding an animal showing a late deployment at 72hr post-dose.

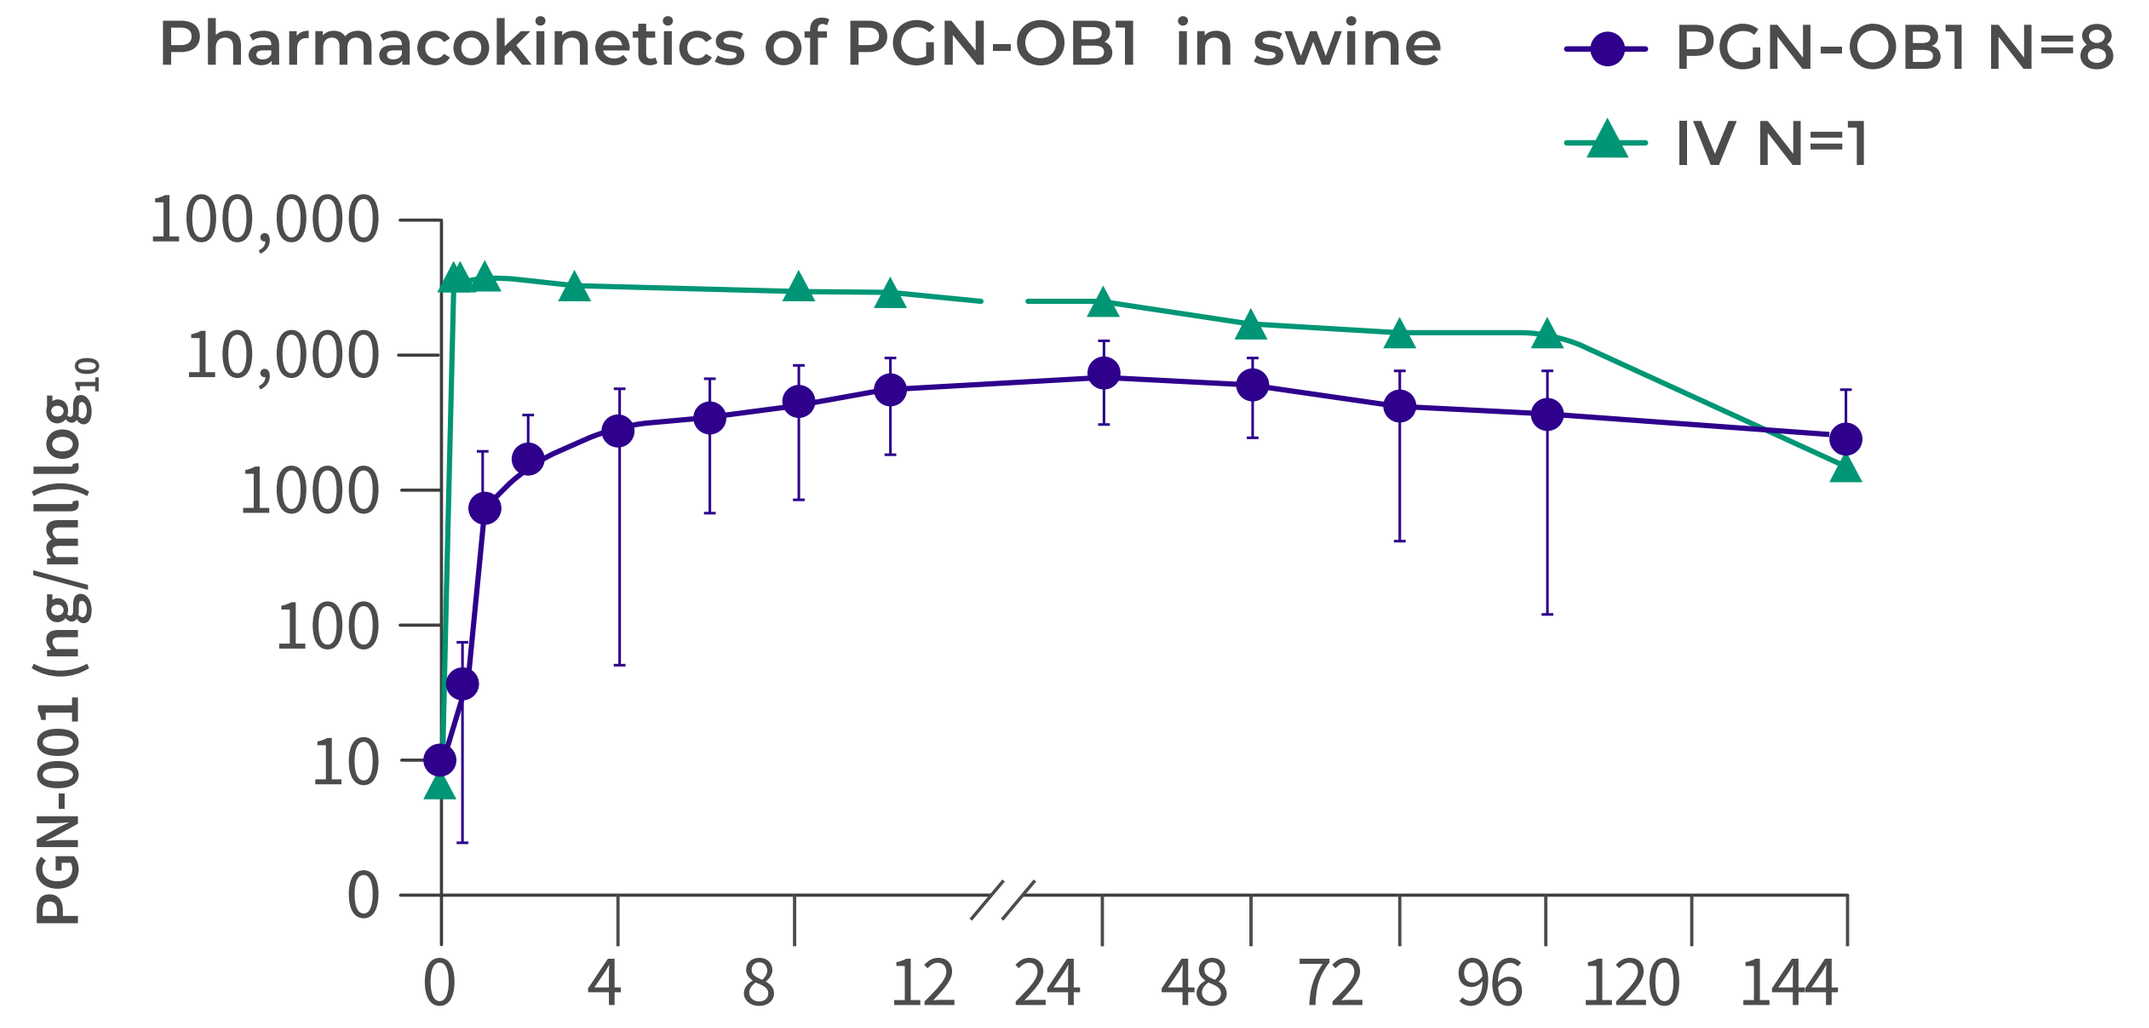
Pharmacokinetics of PGN-OB1 in swine
PGN-OB1 N=8
IV N=1
PGN-001 (ng/ml)log10
100,000
10,000
1000
100
10
0
0
4
8
12
24
48
72
96
120
144

FIGURE 6. Plasma concentration of PGN-OB1 delivered via ID and IV over time

SUMMARY AND DISCUSSION

- Demonstrated ≥ 83% deployment accuracy and consistent deployment time of the OBDS post gastric emptying in the canine small intestine.
- Demonstrated a semi-autonomous OBDS device that can achieve as high as 55% bioavailability in swine, which is a magnitude greater than current oral protein or peptide delivery technology in the market, and at levels much closer to the subcutaneous route of administration estimated in human trials.

© 2022 Biora Therapeutics, Inc. All rights reserved.
Biora Therapeutics™ is a trademark of Biora Therapeutics, Inc.

bioratherapeutics.com

Presented at Parental Drug Association Universe of Pre-Filled Syringes and Injection Devices Conference, October 18-19, 2022

CONTACT INFO
Chris Wahl, MD, MBA
VP, Strategy and Business Development
chris.wahl@bioratherapeutics.com