Results of human device function studies for the NaviCap™ Targeted Oral Delivery Platform in healthy volunteers and patients with UC

Shaoying Nikki Lee,[1] Ghesal Razag,[1] Colleen Kelly,[1] Erik Sandefer,[2] Walter Doll,[2] Richard Page,[2] John Conklin,[1] Paul Shabram,[1] Sharat Singh,[1] and Ariella Kelman[1]

1. Biora Therapeutics, Inc., 2. Scintipharma Inc.

BIORA™ Therapeutics

INTRODUCTION

Despite progress in the treatment of patients with ulcerative colitis (UC), outcomes remain sub-optimal. Research has shown that insufficient drug concentration in colon tissue is a potential cause of low remission rates.[1,2,3] The NaviCap™ platform is an orally ingestible smart pill equipped with Biora's proprietary GITrac™ localization technology that identifies anatomic locations in the gastrointestinal (GI) tract. In the current platform application, the NaviCap device is designed to deliver a liquid drug formulation directly to the colon mucosa, bypassing the upper GI tract. This has the potential to improve efficacy and reduce systemic toxicity by achieving high tissue and low systemic drug concentrations. A Phase 1 clinical trial of BT-600 (the NaviCap device containing tofacitinib) is ongoing. Results from human device function clinical studies of the NaviCap without drug are summarized here.

The NaviCap Device

- The NaviCap device comprises a drug reservoir that houses a liquid formulation of a therapeutic compound and an electronic module **(Figure 1A)**.
- The electronic module houses the localization system, electronics, and the gas cell required for displacing the drug reservoir from the device at the target location **(Figure 1A)**.

Autonomous Localization

- The autonomous localization system identifies different anatomical regions by emitting colored light that interacts with the local GI environment and returns to spatially separated detectors. Measured light levels are analyzed by a software algorithm to detect changes associated with different anatomical features **(Figure 1B)**.
- Upon detection of entry into the colon, the device initiates the gas cell actuator for drug release.

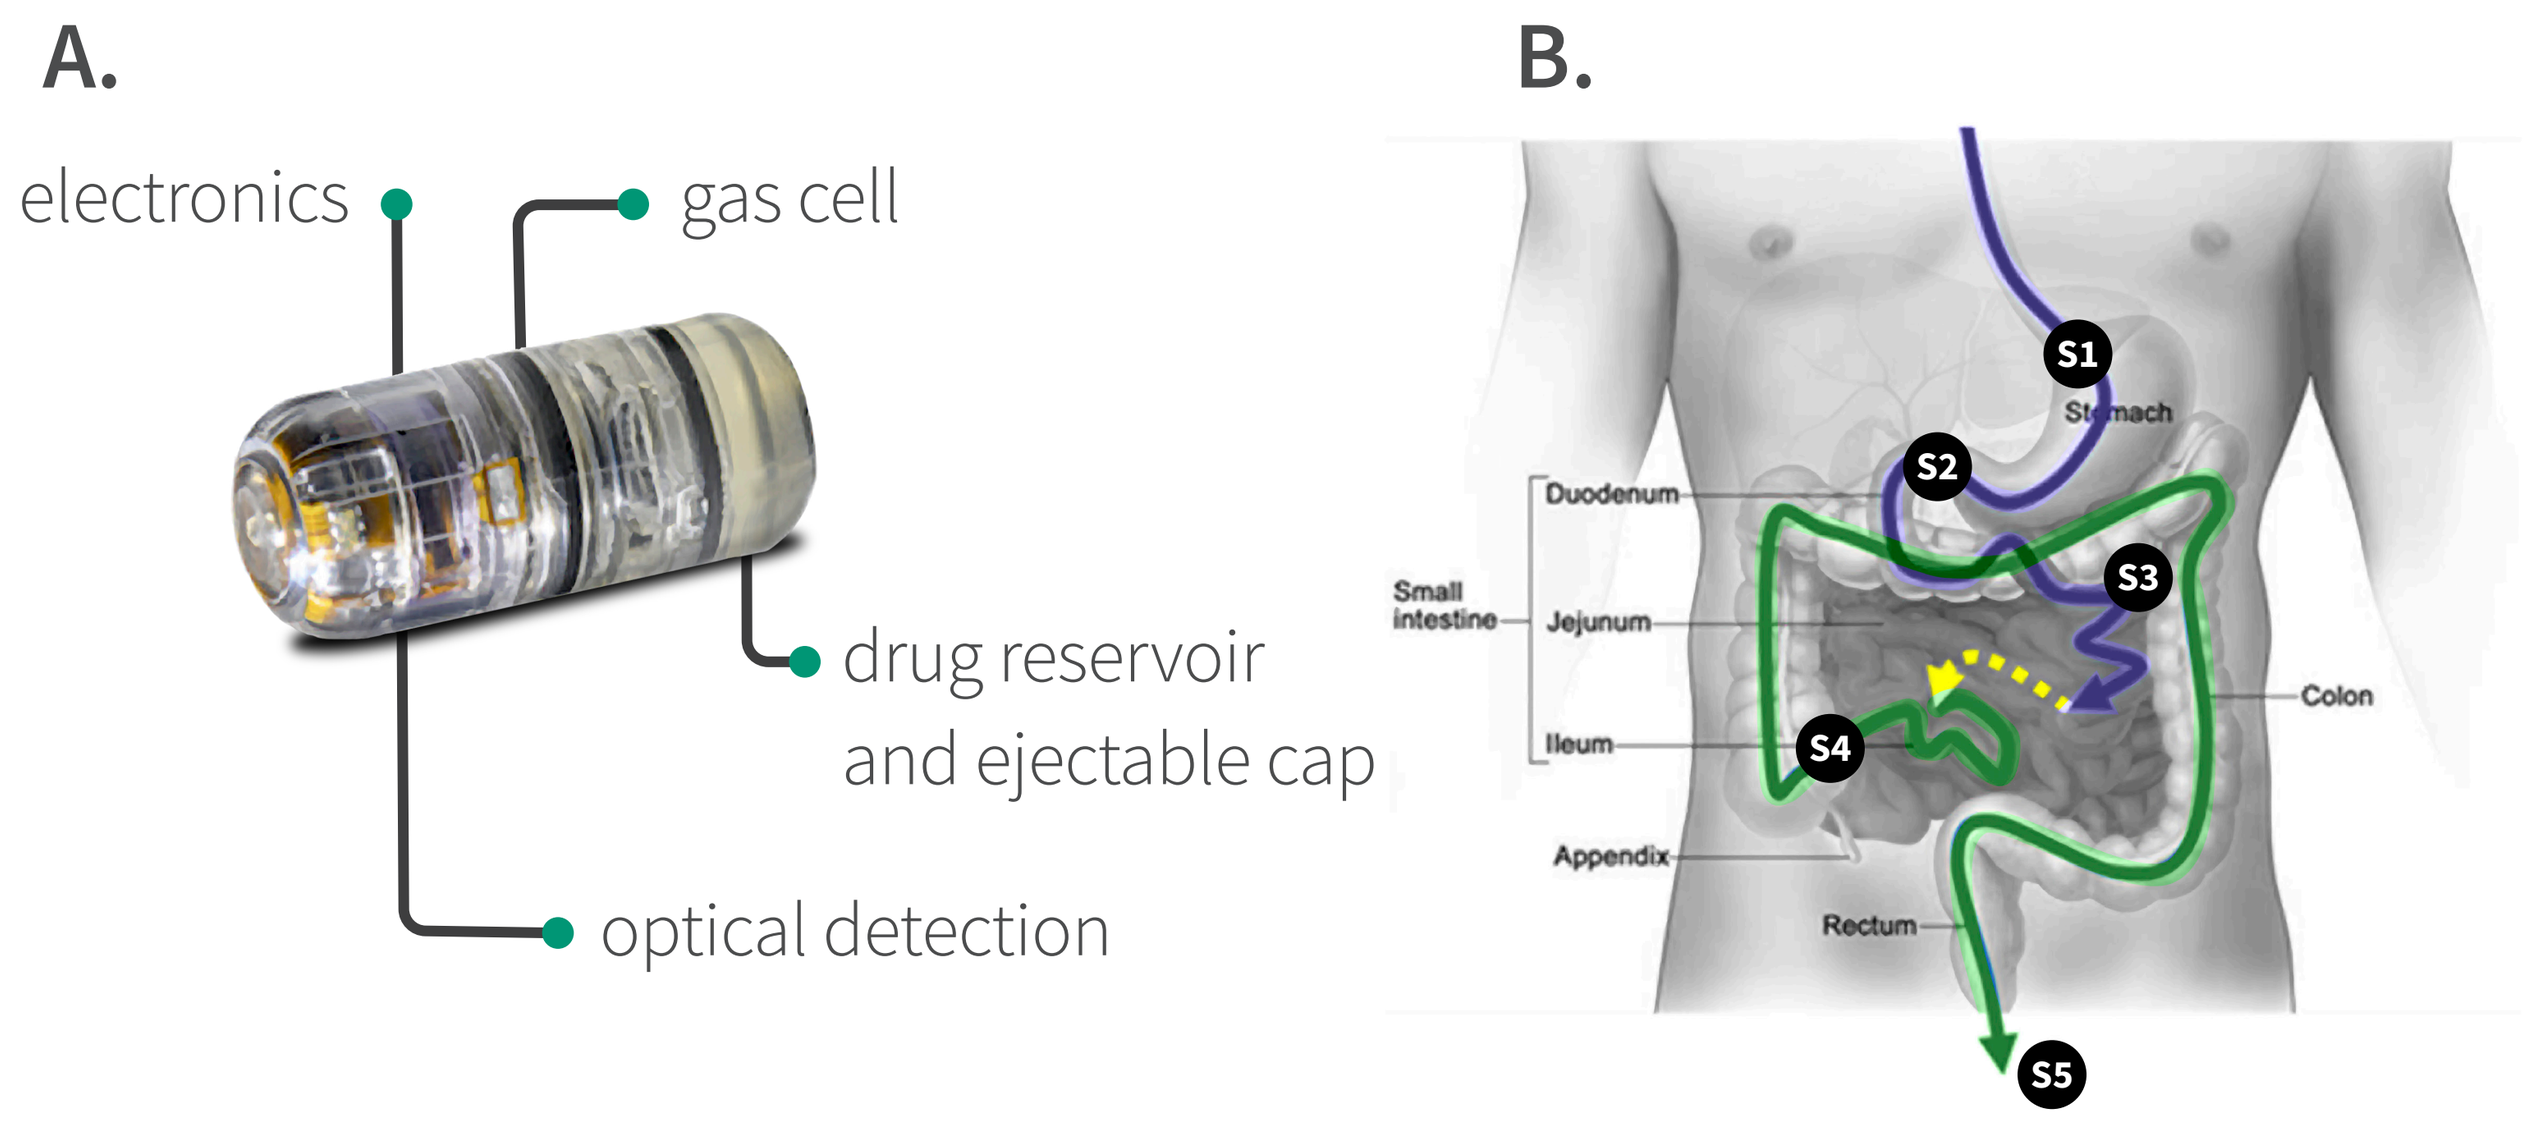
A.
electronics
gas cell
drug reservoir
and ejectable cap
optical detection
B.
S1
Stomach
S2
S3
S4
S5
Duodenum
Small intestine
Jejunum
Ileum
Appendix
Rectum
Colon

FIGURE 1. Autonomous localization technology enables targeted delivery of therapeutics. A. Photograph of the NaviCap device; B. The internal algorithm can detect five major anatomical locations: (S1) entry to the stomach, (S2) entry into the duodenum (gastric emptying), (S3) entry into the jejunum, (S4) entry into the colon, and (S5) exit from the body.

OBJECTIVES

- To evaluate the localization and delivery function of NaviCap devices using gamma scintigraphy in healthy participants and patients with active UC, in a fasted state.
- To assess the safety of NaviCap devices in healthy participants in fasted and fed states and patients with active UC in a fasted state.

METHODS

Four device function studies were performed in human subjects. Three studies used gamma scintigraphy to assess delivery of radiolabeled payload into the colon, and one study assessed device function in both fasted and fed states.

Key Inclusion and Exclusion Criteria

- Male and female participants between 18 and 75 years of age
- Ability to swallow 000 size capsules
- Studies PM-601, PM-611, and BT-603 enrolled healthy participants
- Study PM-602 enrolled patients with active UC confirmed by endoscopy and histology and Mayo score ≥ 2 *or* elevated Fecal Calprotectin Protein *or* elevated high-sensitivity C-reactive protein within one month of the screening visit.

Study Design: Scintigraphy Studies (PM-601, PM-602, BT-603)

- Each study participant was fasted overnight and was administered a single NaviCap device before resuming a normal diet at four hours post-dose.
- Each NaviCap device was filled with radioactive marker indium(In)-111 DTPA to identify device location and to visualize payload release in the GI tract. Water radiolabeled with technetium (Tc)-99m DTPA was co-administered to delineate GI landmarks.
- Device GI transit and delivery location of the radioactive payload were confirmed by serial scintigraphy. Electronic algorithm data from recovered devices was then examined for a signal (S4) indicating colon entry detection. Delivery time and location by scintigraphy was compared with recovered device localization data.

Study Design: Fasted and Fed Study (PM-611)

- Healthy participants were enrolled and administered a single empty NaviCap device weekly in either the fasted state or in one of three possible fed schedules over approximately four weeks.
- Subjects were either fasted overnight and dosed in the morning (fasted schedule) or consumed a light breakfast with equivalent calories and protein/fat content of an egg-beater meal (two large eggs [60 kcal] served with two slices of bread [120 kcal] and 30 g strawberry jam [74 kcal]) and dosed following one of three fed schedules:
 - Fed Schedule 1: Immediately before administration of NaviCap device
 - Fed Schedule 2: 30 minutes post-administration of NaviCap device
 - Fed Schedule 3: 2 hours before administration of NaviCap device
- Electronic algorithm data from the recovered devices was examined for colon entry (S4) signal and activation of payload release.

RESULTS

Adverse Events

The NaviCap device was well-tolerated across 81 administrations in 47 participants, across four human device function studies.

- A total of 40 healthy participants (PM-601 N=12; BT-603 N=16; PM-611 N=12) and 7 patients with active UC (PM-602 N=7) were enrolled.
- In PM-611, mild AEs were reported as possibly related to the device in two participants, including vomiting (N=1) and nausea (N=1), which resolved on the same day.
- In PM-602, one subject with active UC experienced mild intermittent abdominal cramping, assessed as unlikely related to device administration by the investigator, and resolved on the same day.
- No other device-related events were reported.

TABLE 1: Identification of colon entry and payload delivery in scintigraphy studies

Parameter	PM-601 (D = 12)	PM-602 (D = 7)	BT-603 (D = 16)	Combined (D = 35)
Identification of Colon Entry Correct S4 call (%) 95% Confidence Interval	10 (83.3%) (55.2%, 95.3%)	7 (100%) (64.6%, 100%)	15 (93.8%) (71.7%, 98.9%)	32 (91.4%) (77.6%, 97.0%)
Payload Delivery Delivered in colon (%)* 95% Confidence Interval	8 (66.7%) (39.1%, 86.2%)	7 (100%) (64.6%, 100%)	15 (93.8%) (71.7%, 98.9%)	30 (85.7%) (70.6%, 93.7%)

*Payload was delivered in 31 subjects, with 30/31 delivered in colon. (In PM-601, delivery was in small intestine for one subject.)

Scintigraphy Studies (PM-601, PM-602, & BT-603)

The NaviCap device released the radioactive payload (^{111}In-DTPA) directly into the colon regardless of variable GI transit time, the level of inflammation, or the presence of blood in stool.

- A total of 32 of 35 NaviCap devices (91.4%) successfully identified colon entry (designated as S4), and 30 of 35 NaviCap devices (85.7%) delivered the radio payload into the colon **(Table 1)**.
- The payload covered the length of the colon: the dispersion of the ^{111}In-DTPA payload completely covered the colon over time and spread to match the ^{99m}Tc-DTPA water coverage area from the site of release throughout the remainder of the colon **(Figure 2)**.
- NaviCap identified colonic entry and released payload in the colon regardless of variable GI transit across both healthy participants and patients with active UC **(Table 2)**.

Fasted and Fed Study (PM-611)

There was no clear evidence of food effects on NaviCap localization and delivery performance.

- A total of 11 out of 12 enrolled healthy participants completed all four eating schedules.
- Out of 44 devices administered in 11 participants, 43 were recovered following administration, and data was successfully recovered from 39 devices.
 - All 39 devices successfully identified colon entry (designated as S4) and activated gas cells for delivery in all eating schedules (100%) **(Table 3)**.
 - Thirty-eight out of 39 recovered devices (97.4%) successfully activated the payload release function **(Table 3)**.

TABLE 3: Device performance in fasted and fed states (analysis population N=11 subjects)

Parameter	Fasted State (D = 9)*	Fed State (D = 30)*	All (D = 39)*
Identification of Colon Entry S4 call (%) Wilson-Score 95% CI	9 (100%) (70.1%, 100%)	30 (100%) (88.6%, 100%)	39 (100%) (91.0%, 100%)
H2 Cell Activation Activated (%) Wilson-Score 95% CI	9 (100%) (70.1%, 100%)	30 (100%) (88.6%, 100%)	39 (100%) (91.0%, 100%)
Payload Release Activation Delivered (%) Wilson-Score 95% CI	9 (100%) (70.1%, 100%)	29 (96.7%) (83.3%, 99.4%)	38 (97.4%) (86.8%, 99.5%)

*Total number of NaviCap devices with successfully retrieved data

CONCLUSION

- **The NaviCap platform's ability to function across variable GI pH, motility, and eating schedules illustrates its potential capability to deliver therapeutics locally to the colon of patients with UC.**
- **Localized delivery could potentially improve efficacy while minimizing systemic exposure and toxicity that is associated with systemic drug delivery.**

TABLE 2: Comparison of the gastrointestinal transit and motility, location of device at time of S4 call, and release of payload across scintigraphy studies

Study Number	Subject ID	Mayo Score	Gastric emptying time (min)	Small intestine residence time (min)	Arrival time at cecum (min)	Device location at S4 call time	Release time of ^{111}In (min)	Device location at time of release	Device recovery time (hr)	Visible blood in stool	Bowel movements to recover device
PM-602: UC Patients	602-116-001	6	104	169	273	Cecum	323	Cecum	47.67	Yes	3
	602-116-002	3	23	115	138	Cecum	188	Cecum	26.5	No	3
	602-116-003	8	38	285	323	Cecum	353	Cecum	32.83	Yes	5
	602-116-004	2	14	249	263	Ascending Colon/ Splenic Flexure	308	Splenic Flexure	24.25	No	1
	602-116-005	6	9	149	158	Cecum	188	Cecum	6.83	Yes	3
	602-116-006	2	87	498	585	Cecum	616	Cecum/ Ascending Colon	48.67	No	9
	602-116-007	3	465	105	570	Cecum/ Ascending Colon	638	Cecum/ Ascending Colon	23.75	No	2
PM-601 and BT-603: Healthy Participants	Median of all subjects (N = 28)	N/A	44.5	232.5	267	Cecum/Ascending Colon/Splenic Flexure	345	Cecum/Ascending Colon/Splenic Flexure	23.75	No	1 (n = 13) 2 (n = 10) 3 (n = 5)

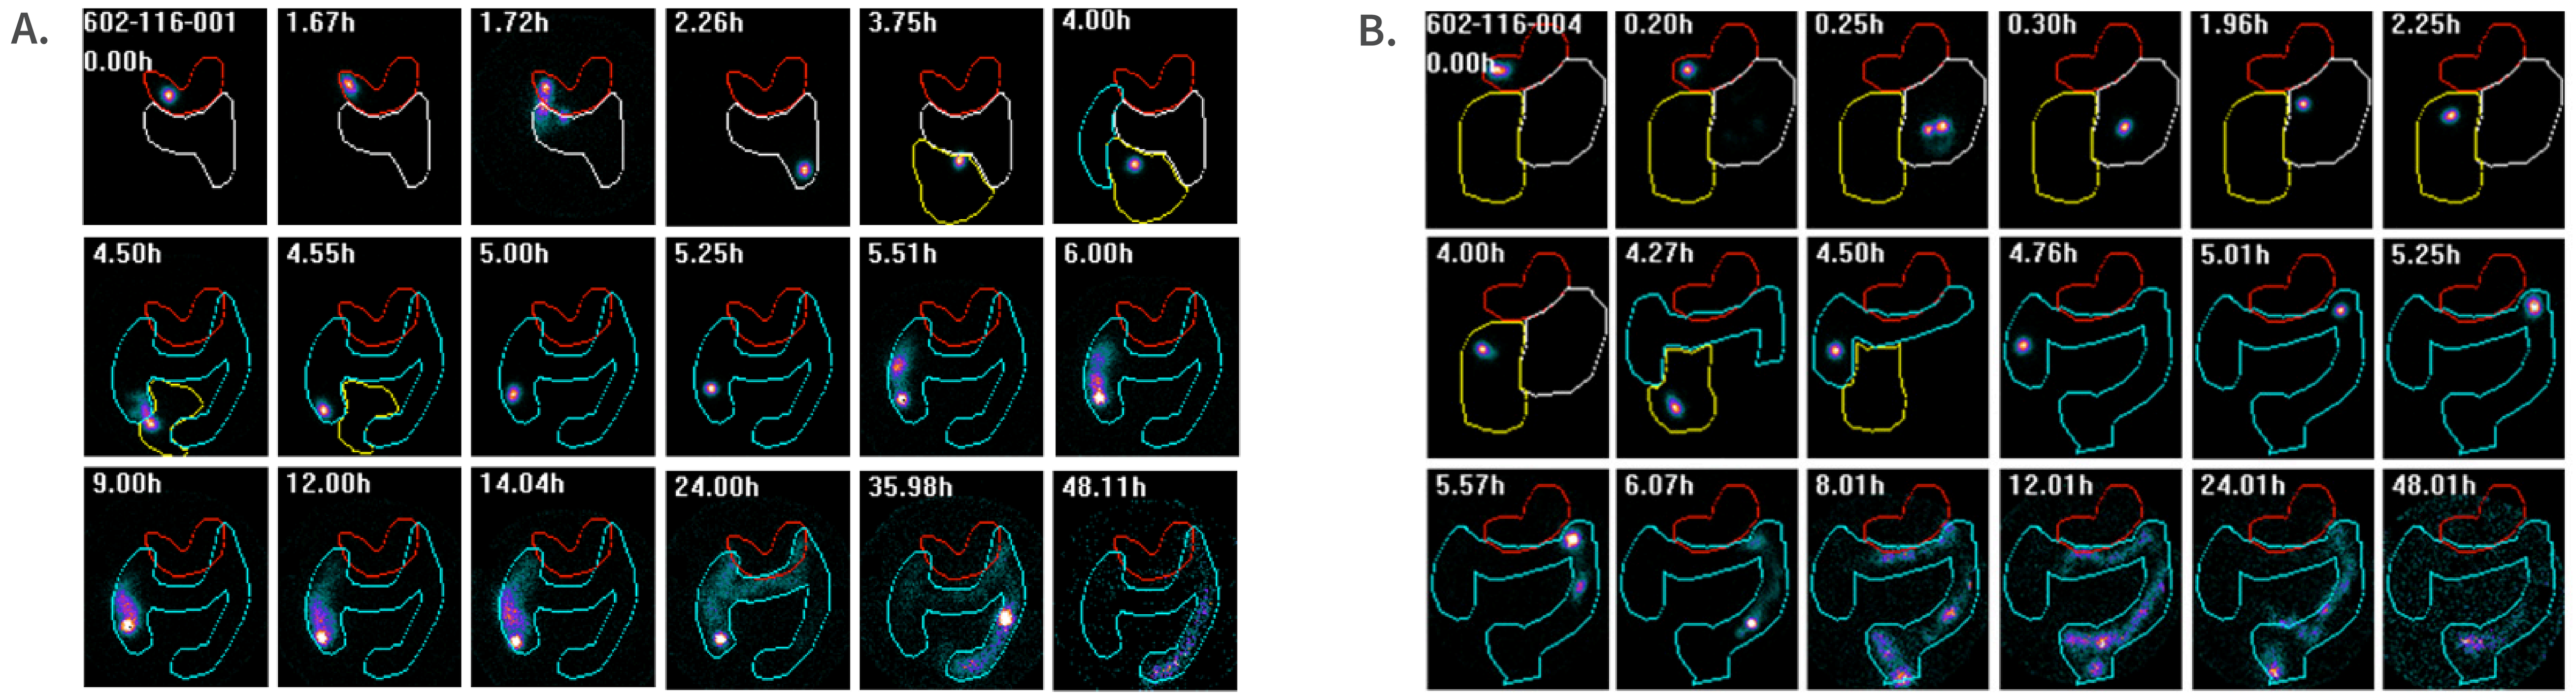
A.
602-116-001
0.00h
1.67h
1.72h
2.26h
3.75h
4.00h
4.50h
4.55h
5.00h
5.25h
5.51h
6.00h
9.00h
12.00h
14.04h
24.00h
35.98h
48.11h
B.
602-116-004
0.00h
0.20h
0.25h
0.30h
1.96h
2.25h
4.00h
4.27h
4.50h
4.76h
5.01h
5.25h
5.57h
6.07h
8.01h
12.01h
24.01h
48.01h

Stomach = Red ROI; Proximal Small Intestine = White; Distal Small Intestine = Yellow; Colon = Cyan

Stomach = Red ROI; Proximal Small Intestine = White; Distal Small Intestine = Yellow; Colon = Cyan

FIGURE 2: Examples of scintigraphy images of cumulative distribution of radiotracer ^{111}In-DTPA release from device post-administration in subjects with active UC and variable disease activity. A. Subject 602-116-001: Mayo score 6, and visible blood in stool. B. Subject 602-116-004: Mayo score of 2 and fast GI motility (gastric emptying in 14 min).

References
1. Verstockt B, Alsoud D, van Oostrom J, et al. Poster presented at: 17th Congress of the European Crohn's and Colitis Organisation (ECCO), February 18, 2022, virtual.
2. Yarur AJ, Jain A, Sussman DA, et al. The association of tissue anti-TNF drug levels with serological and endoscopic disease activity in inflammatory bowel disease: the ATLAS study. Gut. 2016;65(2):249-255.
3. Pauwels RWM, Proietti E, van der Woude CJ, et al. Vedolizumab Tissue Concentration Correlates to Mucosal Inflammation and Objective Treatment Response in Inflammatory Bowel Disease. Inflamm Bowel Dis. 2021;27(11):1813-1820. doi:10.1093/ibd/izab053

Poster #Su1778
Presented at Digestive Disease Week, May 18 – 21, 2024, Washington DC.

© 2024 Biora Therapeutics, Inc. All rights reserved. Biora Therapeutics™ is a trademark of Biora Therapeutics, Inc.

bioratherapeutics.com

Contact Info
Shaoying Nikki Lee, PhD
Director, Clinical and Translational Science
nikki.lee@bioratherapeutics.com

SCAN ME

Scan the code to view an electronic version of this poster, or visit **bioratherapeutics.com/publications**